26th ECCMID Amsterdam, Netherlands 9 – 12 April 2016

EV0626

Cellular pharmacokinetics of gepotidacin (GSK2140944) in human THP-1 monocytes (THP-1) and murine macrophages (J774)

F. Peyrusson, F. Van Bambeke, P.M. Tulkens
Louvain Drug Research Institute, *Université catholique de Louvain*, Brussels, Belgium

Mailing address:
P.M. Tulkens
av. Mounier 73
(B1.73.05)
1200 Brussels, Belgium
tulkens@facm.ucl.ac.be
+32-2-762-2136

Background and Aims

Staphylococcus aureus remains a major cause of community and nosocomial infections because of acquired resistance mechanisms (1) and ability to take refuge intracellularly (2). Novel antibiotics, therefore, need to be assessed for activity against resistant strains, including after phagocytosis by eukaryotic cells.

Gepotidacin is a novel triazaacenaphthylene antibiotic which inhibits bacterial type IIA topoisomerases (Figure 1). We previously showed that gepotidacin is active against intracellular forms of *S. aureus*, including strains resistant to other antibiotics, in human THP-1 monocytes (3).

The goal of the present study was to characterize the cellular pharmacokinetics of gepotidacin in human monocytes (THP-1; undifferentiated cells growing in suspension) and murine macrophages (J774; differentiated and adherent).

Figure 1: Structure of gepotidacin (GSK2140944)

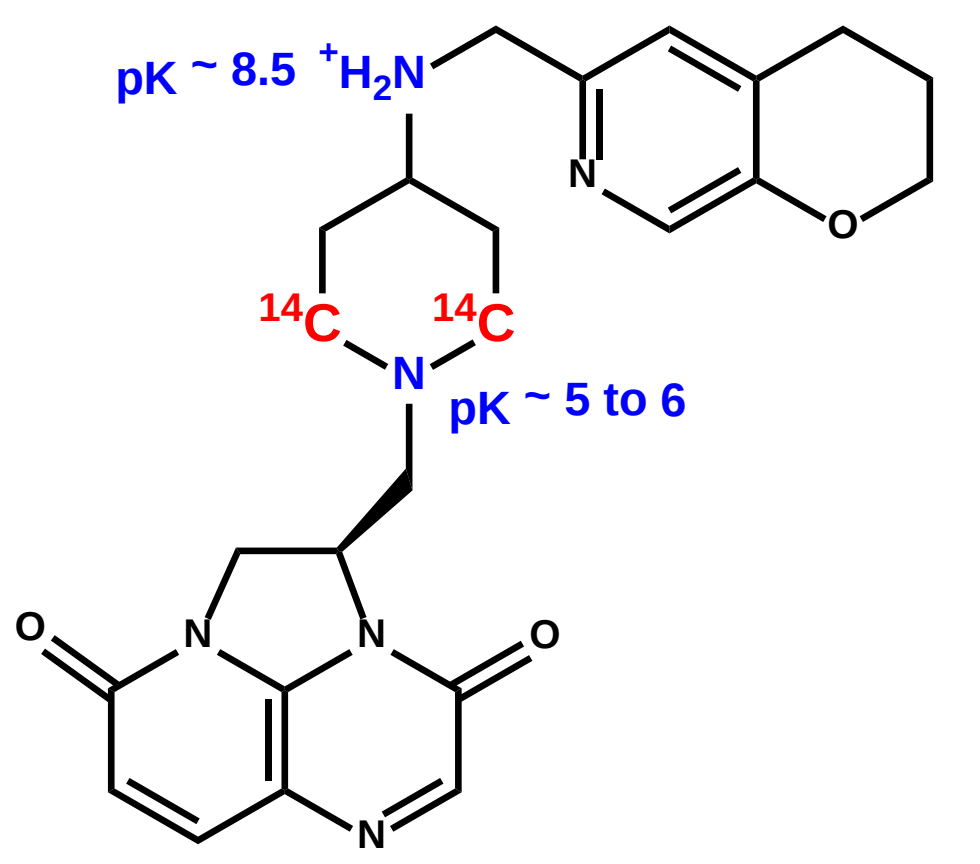
pK ~ 8.5 ⁺H2N
14C
14C
N
pK ~ 5 to 6
O
N

The figure shows

(i) the positions of the labelled atoms (^{14}C)

(ii) the calculated pKa's of the two aminofunctions (in **blue**) assumed to be ionizable in the 5-8 pH range (predominant microspecies: monoionic [68%] at pH 7.0) and dicationic [93%] at pH 5.5).

Sources: structure redrawn from [5]; physicochemical constants: ChemBioDraw (http://www.cambridgesoft.com/) and Reaxys® Reed Elsevier (http://www.reaxys.com)

Methods

Cell lines: murine J774 macrophages (expressing P-gp and MRP efflux transporters) and human THP-1 monocytes

Gepotidacin: [^{14}C]-gepotidacin and unlabelled compound (99.7% purity).

Accumulation and efflux studies: Cells incubated with gepotidacin and collected at the end of the experiment after washing for measurement of (i) cell-associated radioactivity and (ii) total protein content (cell volume: 3.08 and 5 µL/mg of cell protein for J774 macrophages (4) and THP-1 cells, respectively). Verapamil and gemfibrozil were used as inhibitors of P-glycoprotein (P-gp) and Multidrug Resistant Protein (MRP) eukaryotic efflux transporters (4,5).

Bacterial strain and cell infection: ATCC 25923 (fully susceptible MSSA; gepotidacin MIC = 0.5 mg/L) used as depicted in Figure 2 (6). Gepotidacin was used at 0.1 mg/L to avoid killing intracellular bacteria.

Figure 2: Cell infection

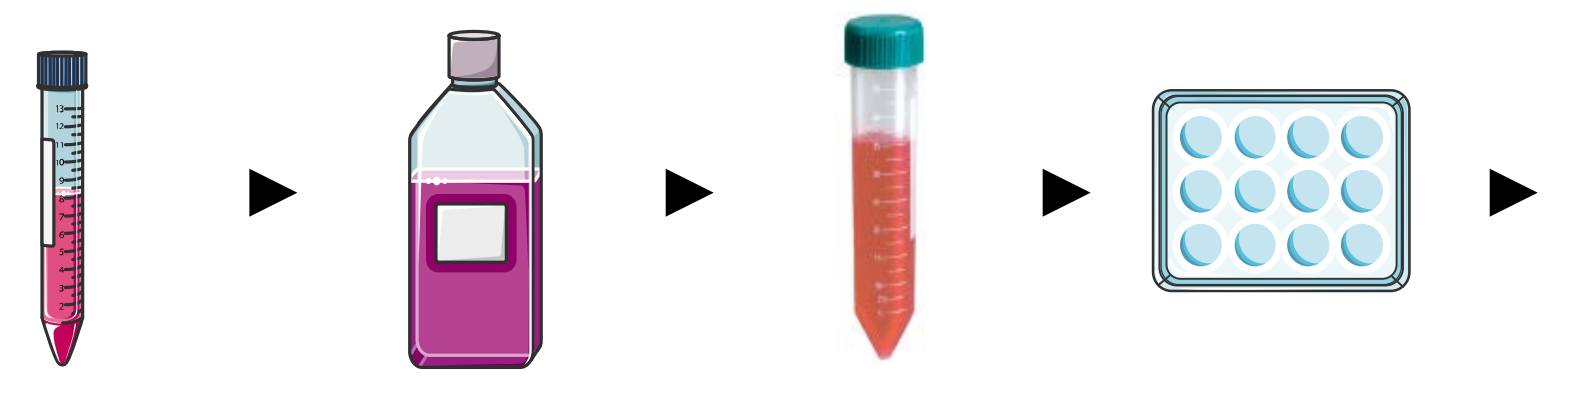

opsonization (human serum) ▶ **phagocytosis** (1 h – 37°C) ▶ **removal of extracellular bacteria** (gentamicin) ▶ **Incubation** • 2 h – 37°C • 30 min with gepotidacin ▶

- **cell collection and homogenization**
- **CFU counting and assay of cell-associated radioactivity and proteins**

Results

Figure 3: Kinetics of accumulation and efflux (0-60 min) and effect of concentration

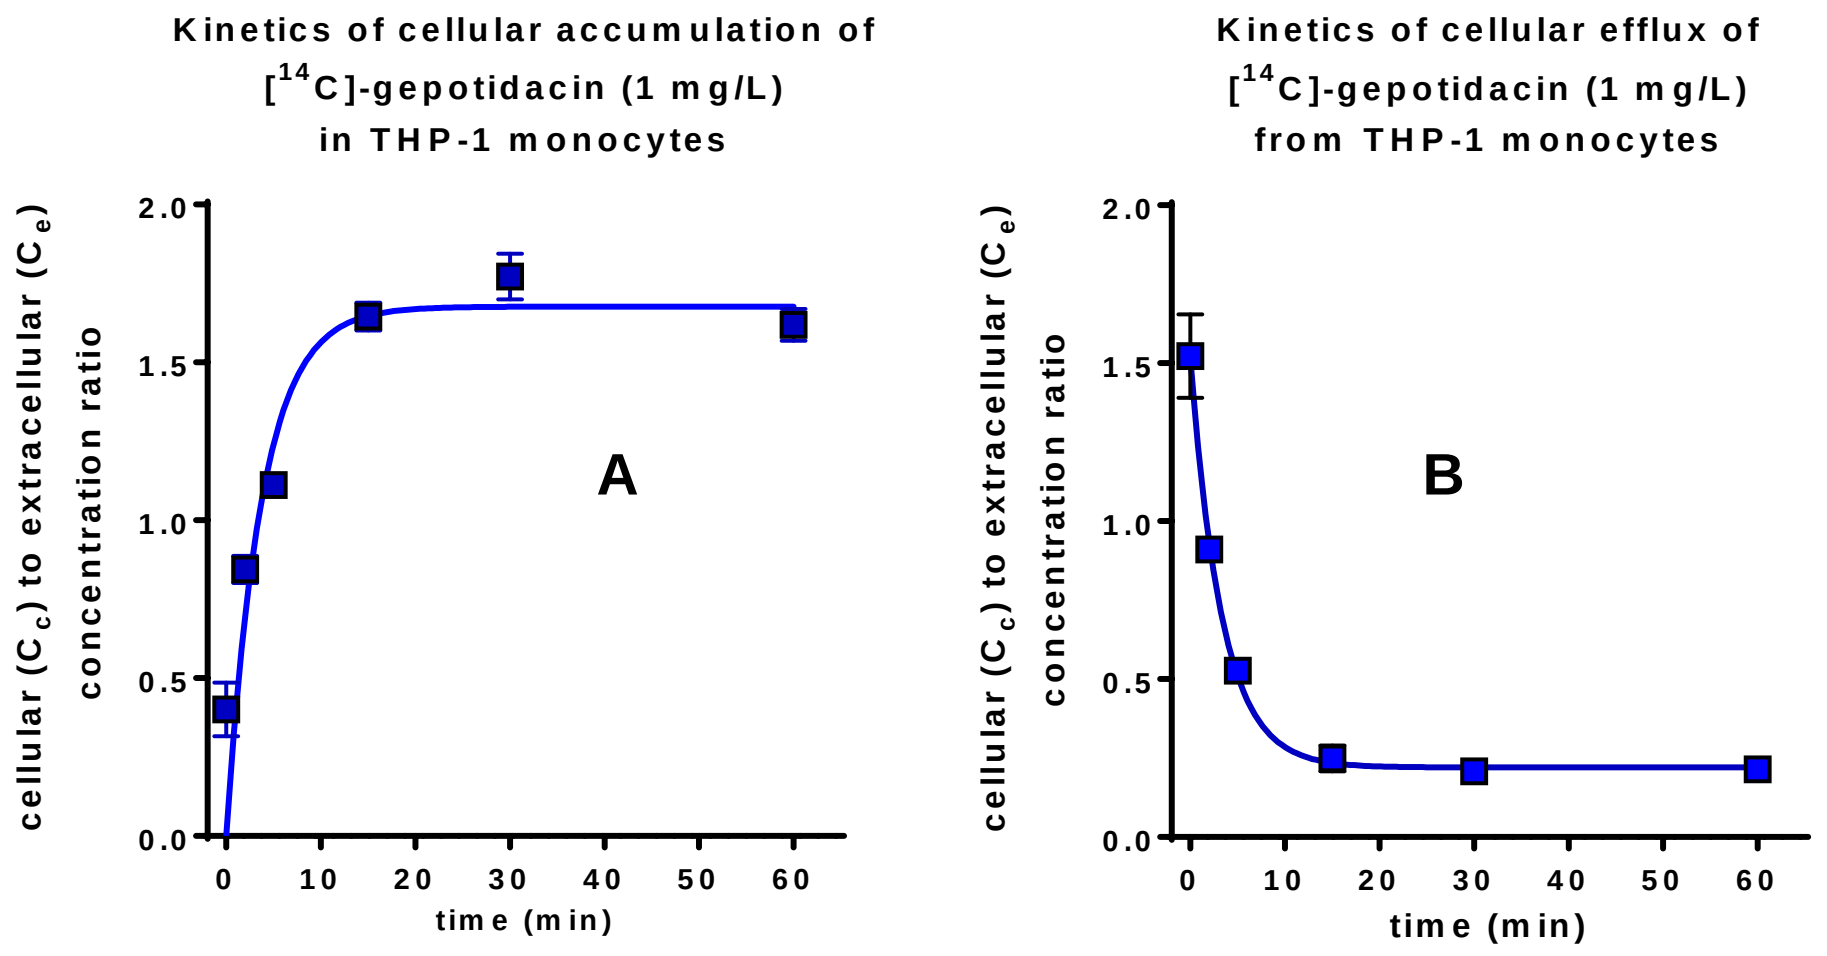
Kinetics of cellular accumulation of [14C]-gepotidacin (1 mg/L) in THP-1 monocytes
Kinetics of cellular efflux of [14C]-gepotidacin (1 mg/L) from THP-1 monocytes
cellular (Cc) to extracellular (Ce) concentration ratio
time (min)
A
B

Influx (A)
- k=0.27±0.06 min^{-1}
- $t_{1/2}$=2.5 min (CI=1.7 to 4.9)

Efflux (B)
- k=0.30±0.02 min^{-1}
- $t_{1/2}$=2.3 min (CI=1.9 to 2.8)

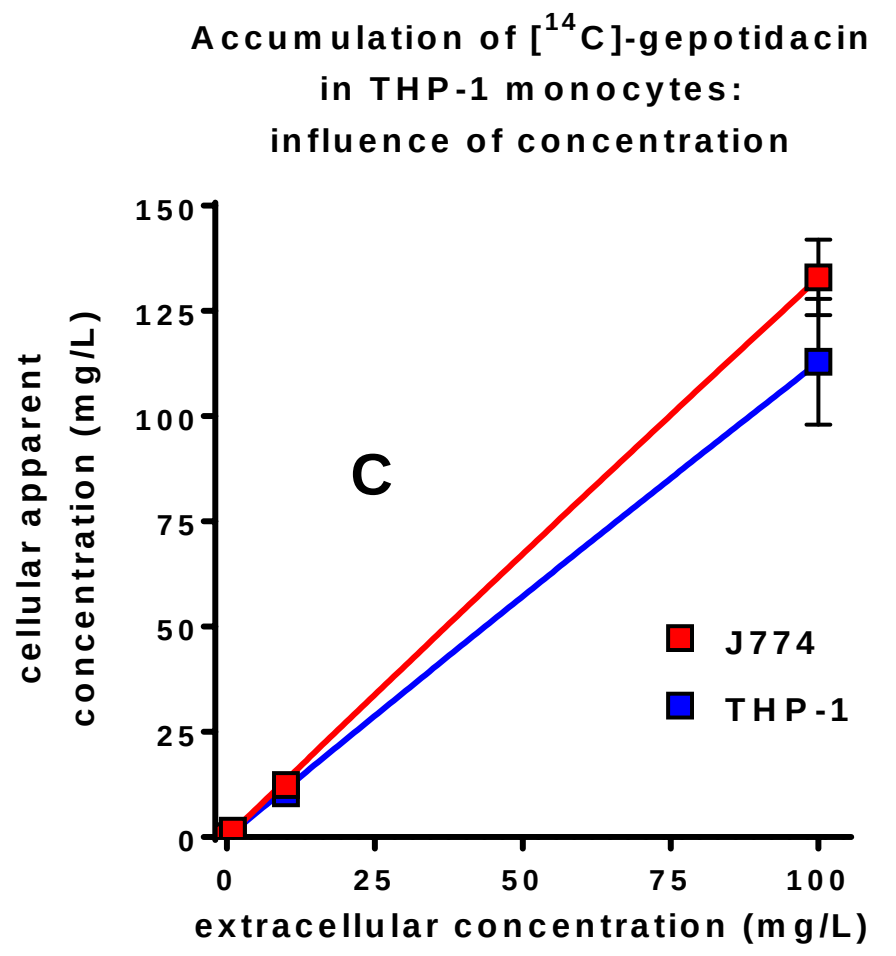
Accumulation of [14C]-gepotidacin in THP-1 monocytes: influence of concentration
cellular apparent concentration (mg/L)
extracellular concentration (mg/L)
C
J774
THP-1

Fast in and out fluxes (equilibrium after 15 min [A] and [B]) and non-saturable accumulation [C]

Figure 4: Influence of temperature and infection on accumulation and efflux

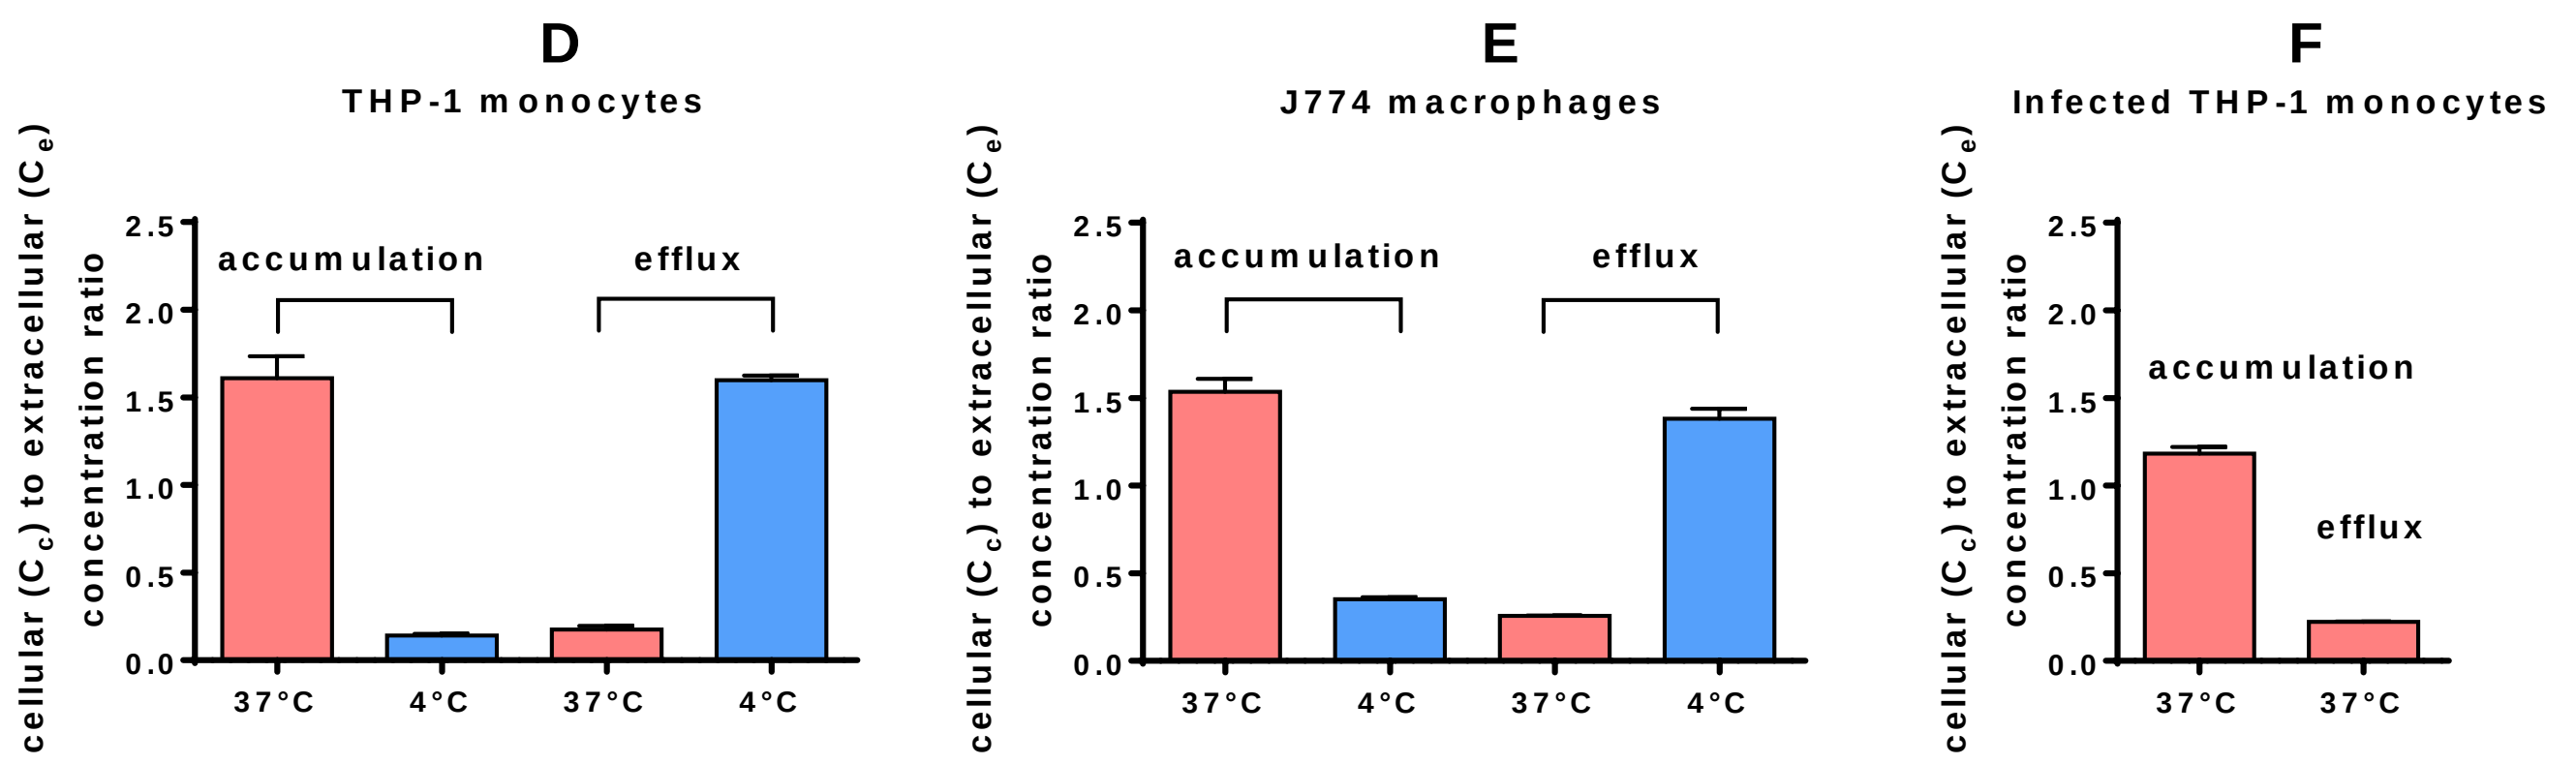
D
THP-1 monocytes
E
J774 macrophages
F
Infected THP-1 monocytes
cellular (Cc) to extracellular (Ce) concentration ratio
accumulation
efflux
37°C
4°C

- **Accumulation:** incubation at 37 or 4°C with gepotidacin for 30 lin and cell collection
- **Efflux:** 30 min incubation with gepotidacin at 37°C followed by 30 min incubation in gepotidacin-free medium at 37 or 4°C.

Complete efflux at 37°C -- No accumulation nor efflux at 4°C (D,E) -- Minor effect of infection (F)

Figure 5: Influence of eukaryotic efflux inhibitors on accumulation at 37°C

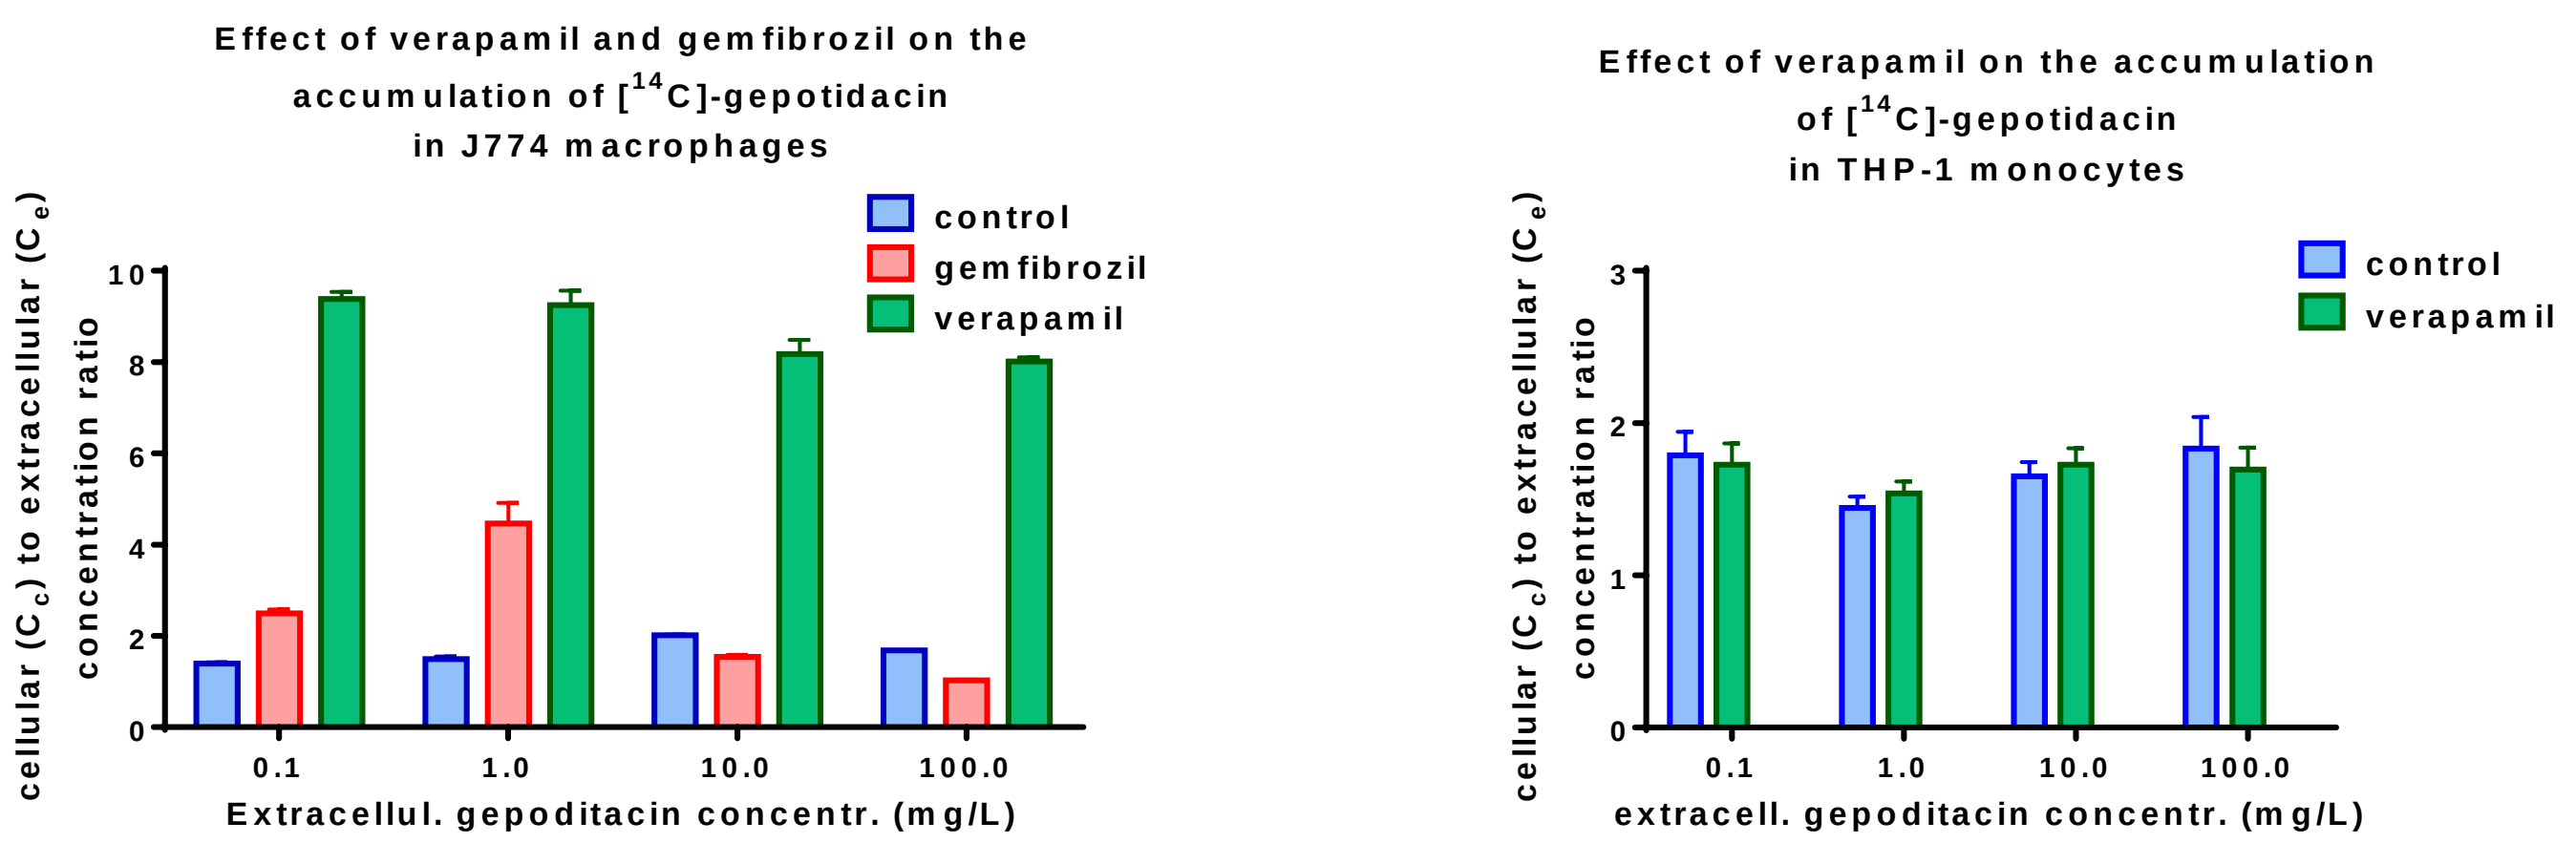
Effect of verapamil and gemfibrozil on the accumulation of [14C]-gepotidacin in J774 macrophages
control
gemfibrozil
verapamil
cellular (Cc) to extracellular (Ce) concentration ratio
Extracellul. gepoditacin concentr. (mg/L)
Effect of verapamil on the accumulation of [14C]-gepotidacin in THP-1 monocytes
control
verapamil
extracell. gepoditacin concentr. (mg/L)

Cells preincubated for 30 min with verapamil (100 µM) or gemfibrozil (500 µM) of the vehicle [control]) and thereafter for 30 min with gepotidacin and the inhibitors (or the vehicle)

- J774 macrophages: marked increase in accumulation by verapamil (P-gp-inhibitor) but modest effect of gemfibrozil (MRP-inhibitor) at concentrations ≤ 1 mg/L.
- THP1 monocytes : no significant effect of verapamil

References

1. Hawkey & Jones (2009) J Antimicrob Chemother 64 Suppl 1:i3-10. PubMed: 19675017.
2. Garzoni & Kelley (2009) Trends Microbiol 17:59-65. PubMed: 19208480
3. Peyrusson *et al.* (2015) 54th ICAAC, San Diego: poster 1099
4. Michot *et al.* (2004) Antimicrob Agents Chemother 48:2673-82. PubMed: 15215125
5. Seral *et al.* (2003) Antimicrob Agents Chemother 47:1047-51. PubMed: 12604540
6. Barcia-Macay *et al.* (2006) Antimicrob Agents Chemother 50:841-51. PubMed: 16495241

Funding

This work was supported by GSK, Collegeville, USA and has been funded in whole or in part with Federal funds from the Office of the Assistant Secretary for Preparedness and Response, Biomedical Advanced Research and Development Authority, under Contract No. HHSO100201300011C. F.P. is paid by the *Université catholique de Louvain*, F.V.B. is Senior Research Associate of the Belgian *Fonds de la Recherche Scientifique* (F.R.S.-FNRS).

Discussion and Conclusions

- The kinetic data suggest that gepotidacin enters the cells by passive diffusion (rapid influx and efflux; no saturation), which probably allows it to be bioavailable and to exert its antibacterial activity intracellularly, as demonstrated with *S. aureus* (3).
- Further experiments will need to assess intracellular localization of gepotidacin in both non-infected and infected cells and to measure its ability to reach its bacterial target.

This poster will be made available after the meeting at http://www.facm.ucl.ac.be/posters