ECCMID 2012
P1602

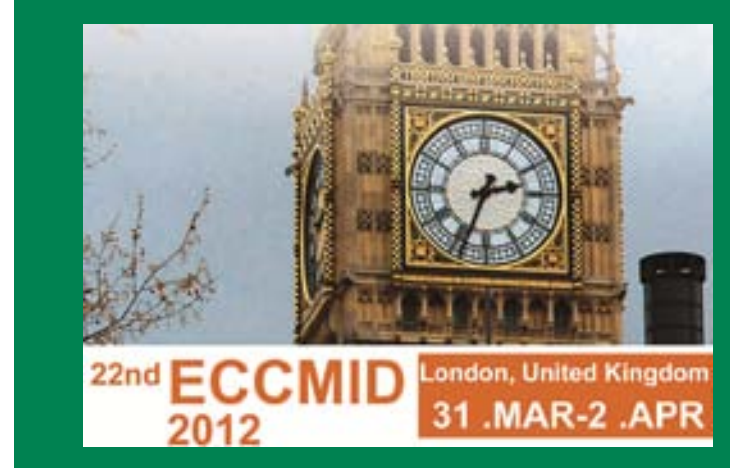

Pharmacodynamic Analysis of the Susceptibility of Intracellular Methicillin-susceptible, and Methicillin-, Vancomycin (VAN)-, and Linezolid (LZD)-resistant *Staphylococcus aureus* to Ceftaroline (CPT)

A. Melard, S. Lemaire, P.M. Tulkens, F. Van Bambeke

Pharmacologie Cellulaire et Moléculaire, Louvain Drug Research Institute
Université Catholique de Louvain, Brussels, Belgium

Paul M. Tulkens
av. Mounier 73,B1.73.05
1200 Brussels – Belgium
tulkens@facm.ucl.ac.be

Abstract (revised)

Objectives:
S. aureus is a facultative intracellular pathogen, explaining the persistent and recurrent character of staphylococcal infections. Ceftaroline (CPT; active metabolite of the prodrug ceftaroline fosamil), is a new broad spectrum cephalosporin with activity against *S. aureus* resistant to methicillin (MRSA) and linezolid (LZD^R). Our aim was to quantitatively assess the activity of CPT against intracellular forms of *S. aureus* using strains with different susceptibilities to CPT and LZD.

Methods:
***Strains**:* Nine strains with MICs (mg/L; pH 7.4) from 0.125 to 2 for CPT, 0.5 to 4 for VAN, and 1 to 8 for LZD; MICs determined at pH 7.4 and 5.5 (to mimic plasma and intracellular environments, respectively) in MH broth (microdilution).
***Cells**:* human THP-1 macrophages (unstimulated).
***Phagocytosis**:* opsonized bacteria (45 min; fresh human serum) at a bacteria:cell ratio of 4 (1 h; 37°C), and elimination of non-internalized bacteria by washing and 45-min incubation with 50 mg/L gentamicin.
***Assessment of activity**:* Incubation of cells (24 h) with CPT (0.01 to 100 mg/L), followed by cell collection (centrifugation and washing in PBS) and enumeration of cell-associated bacteria (CFU/mg cell protein). Results are expressed as the change in intracellular inoculum at 24 h compared to time 0, with data used to fit a Hill equation of the dose-response for determination of static concentration (C_s) and maximal relative efficacy (E_{max}).

Results:
The MICs of CPT towards MRSA were systematically 1 to 2 dilutions lower at pH 5.5 vs. pH 7.4. In concentration-response experiments, (i) E_{max} was between -0.3 and -0.8 log_{10} CFU decrease compared to post-phagocytosis inoculum; (ii) strains with a higher MIC (broth pH 7.4) showed a trend (not significant) toward correspondingly higher C_s, with the highest value, however, still remaining < C_{max}; (iii) all strains behave alike when data are plotted against multiples of MIC at pH 5.5, with a C_s about 8-fold larger than MIC at that pH; a VISA and, to a lesser extent, a LZD resistance phenotype was without effect on CPT activity.

Conclusions:
CPT is active against intraphagocytic *S. aureus* (disregarding their resistance phenotype to beta-lactams, VAN or LZD), for MICs (broth pH 7.4) up to 2 mg/L. The approximately 8-fold higher difference between the C_s and MIC at acid pH may be a function of the kinetics of CPT uptake or reflect effects of the intracellular environment on CPT activity.

Introduction

Methicillin-resistant *Staphylococcus aureus* (MRSA) is a major pathogen causing infections for which treatment remains challenging due to (i) increased resistance to several classes of antimicrobial agents (including vancomycin [VAN]) and (ii) intracellular survival within eukaryotic cells.[1] The latter organisms are especially difficult to eradicate since the intracellular environment seems to confer a protective niche from the lethal action of immune defenses and antibiotics.[2] In this context, the novel cephalosporin antibiotic, ceftaroline (CPT; active metabolite of the prodrug ceftaroline fosamil) has a broad spectrum of activity against Gram positive and Gram negative organisms, and is active against MRSA (due to its high affinity for penicillin-binding-protein PBP 2a), including strains with the VISA phenotype or resistant to linezolid (LZD). Little is known, however, about the activity of CPT against intracellular *S. aureus*.

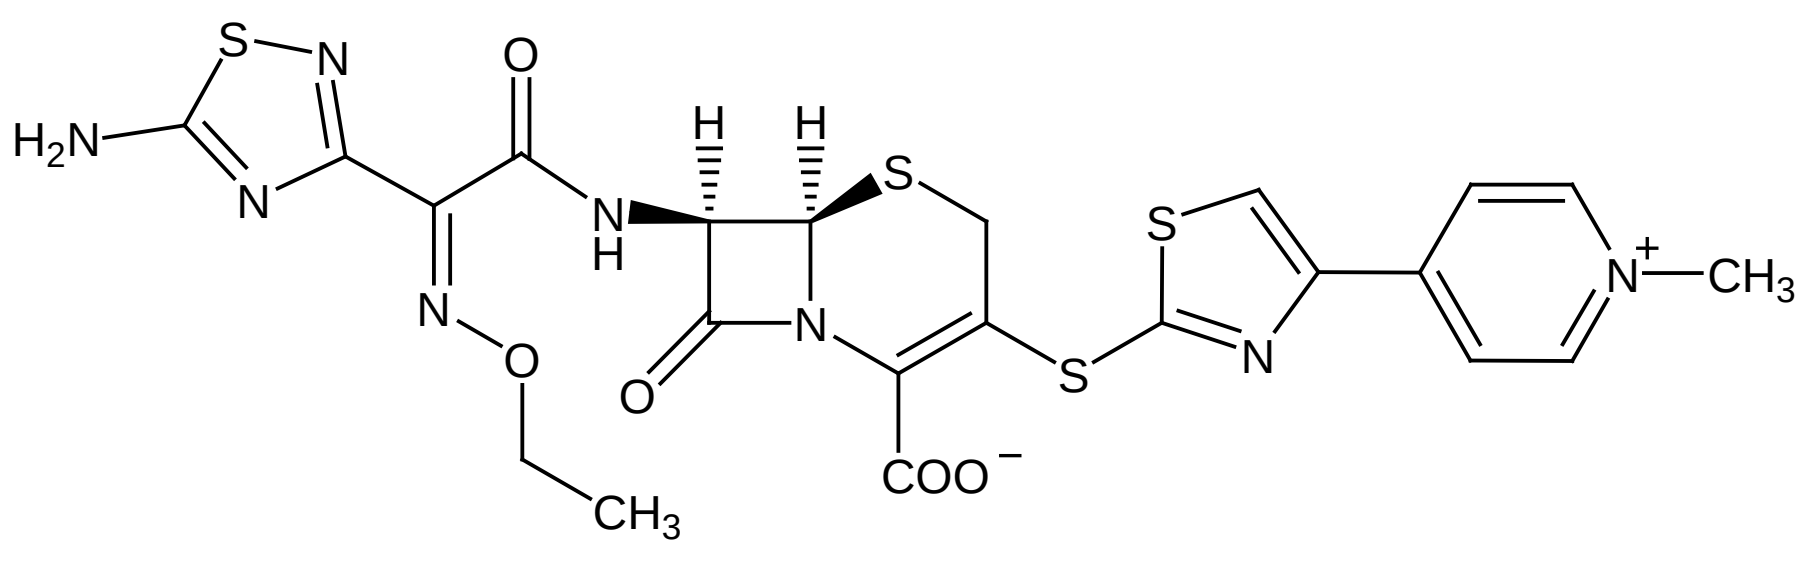

ceftaroline

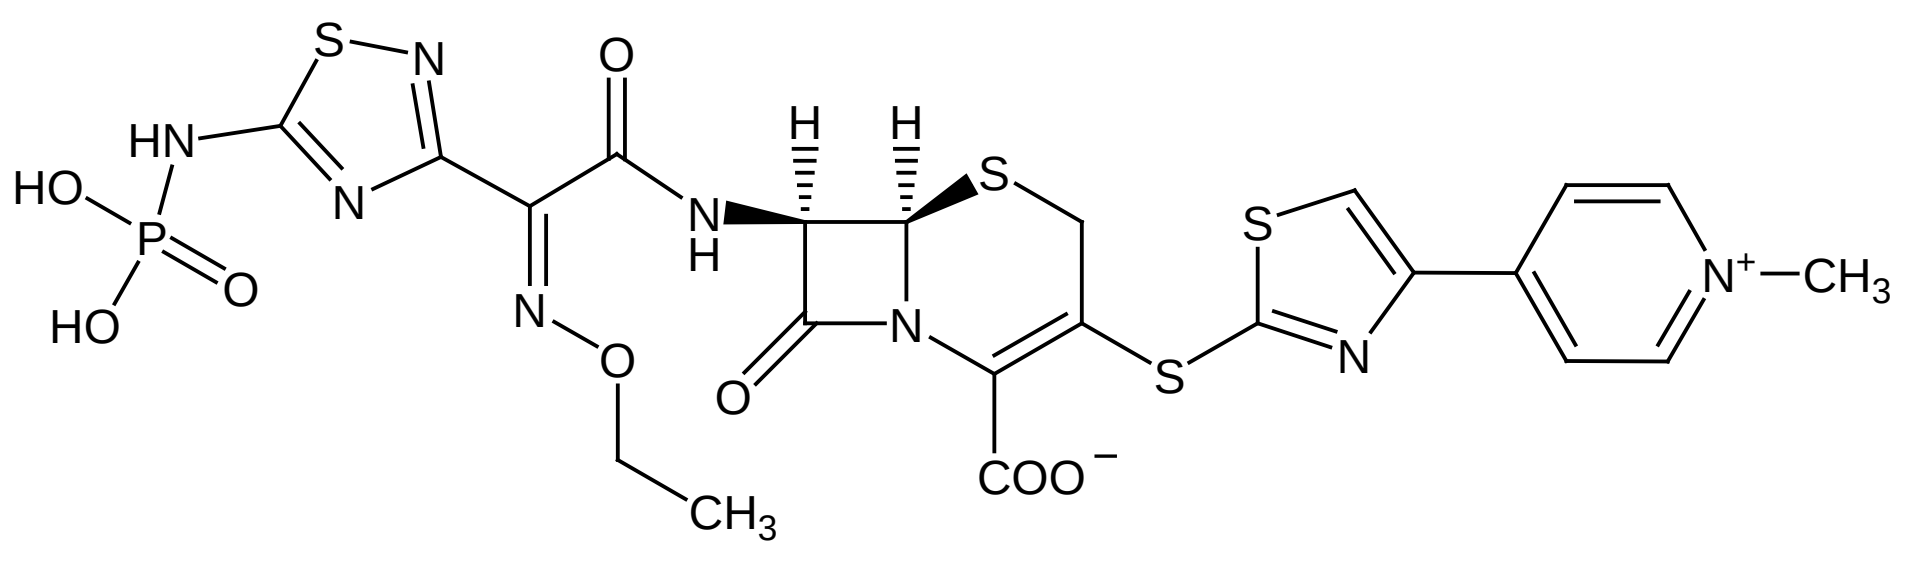

ceftaroline fosamil

Objectives

We aimed at investigating:

(i) the susceptibility of clinically relevant *S. aureus* isolates (incl. multi-resistant strains) to CPT

(ii) the efficacy of CPT towards intracellular forms of these isolates in comparison with VAN and LZD

Methods

Bacterial isolates: The *S. aureus* strains (N=12) were obtained as indicated in Table 1. MICs were determined in MH broth at both pH 7.4 and pH 5.5 to mimic the extracellular and intracellular environments, respectively.

Cells and Cell infection: Human THP-1 monomyelocytic cells were incubated with opsonized bacteria (45 min; fresh human serum) at a bacteria:cell ratio of 4 (1 h; 37°C). Non-internalized bacteria were then removed by washing and 45-min incubation with gentamicin (50 mg/L). Cells were resuspended in cell culture medium (RPMI 1640) in the presence of increasing concentration of antibiotics. After 24 h, cells were harvested by centrifugation, washed with PBS, and lysed (water). Cell lysates were used for enumeration of cell-associated bacteria (CFU) and protein content. Results are expressed as the change in intracellular inoculum at 24 h compared to time 0, with data used to fit a Hill equation of the dose-response for determination of static concentration (C_s), maximal relative efficacy (E_{max}), and concentration yielding a response half-way between E_{max} and absence of effect (E_{min}; unimpaired bacterial growth). Additional details are available in refs 3 and 4.

Results

Table 1. Strains: Phenotypes, Origin and MICs

Strains	Phenotypes and origin	MIC (mg/L) *			
		CPT		VAN	LZD
		pH 7.4	pH 5.5	pH 7.4	pH 7.4
ATCC 25923	MSSA [1]	0.125	0.125	0.5	1
34843	MSSA [2]	0.25	0.125–0.25	1	4
36065	MRSA [2]	0.5	0.125–0.25	2	4
19210	MRSA [2]	0.5	0.25	1	2
ATCC 33591	HA-MRSA [1]	0.5	0.25	0.5	1-2
SA 1984	MRSA [3]	0.5	0.25	0.5	1
SA 555	MRSA-VISA [3]	0.5	0.125	4	0.5
35165	MRSA [2]	1	0.25	0.5	4
062-13101A	MRSA [2]	2 (1.5) [#]	0.5	1	2
062-13091A	MRSA [2]	2	1	1	1-2
48046	MRSA [2]	2	0.5	1	2
CM05	MRSA – LZD^R [4]	2	0.5	1-2	4-8

* Duplicate determinations (with individual values if different)
[#] Arithmetic dilutions
[1] Obtained from the American Type Cell Collection (Manassas, VA)
[2] Obtained from JMI Laboratories, North Liberty, Iowa
[3] Obtained from P.C. Appelbaum (Hershey Medical center, PA)
[4] Obtained from Dr J.P. Quinn: Pfizer Global Research and Development, Groton, Connecticut

MICs ranged between 0.125 to 2 (CPT), 0.5 to 4 (VAN) and 0.5 to 8 (LZD).
When tested at pH 5.5, CPT MICs for MRSA were 1 to 4 log_2 dilutions lower than at pH 7.4 (Table 1).

E_{max} was between -0.23 and -1.03 log_{10} CFU decrease compared to post-phagocytosis inoculum (similar to what was previously observed with ceftobiprole).[4] Strains with a MIC > 1 showed a trend toward a higher C_s, with the highest value still, however, lower than C_{max} (20 mg/L).[5] All strains behave alike when data are plotted against multiples of MIC at pH 5.5 with C_s systematically higher (4.4 to 17 x) than the MIC at that pH (Figures 1a and 1b).

Intracellular Activity

Figure 1a. Comparison of CPT Activity Between MRSA, VISA and LZD^R *S. aureus*

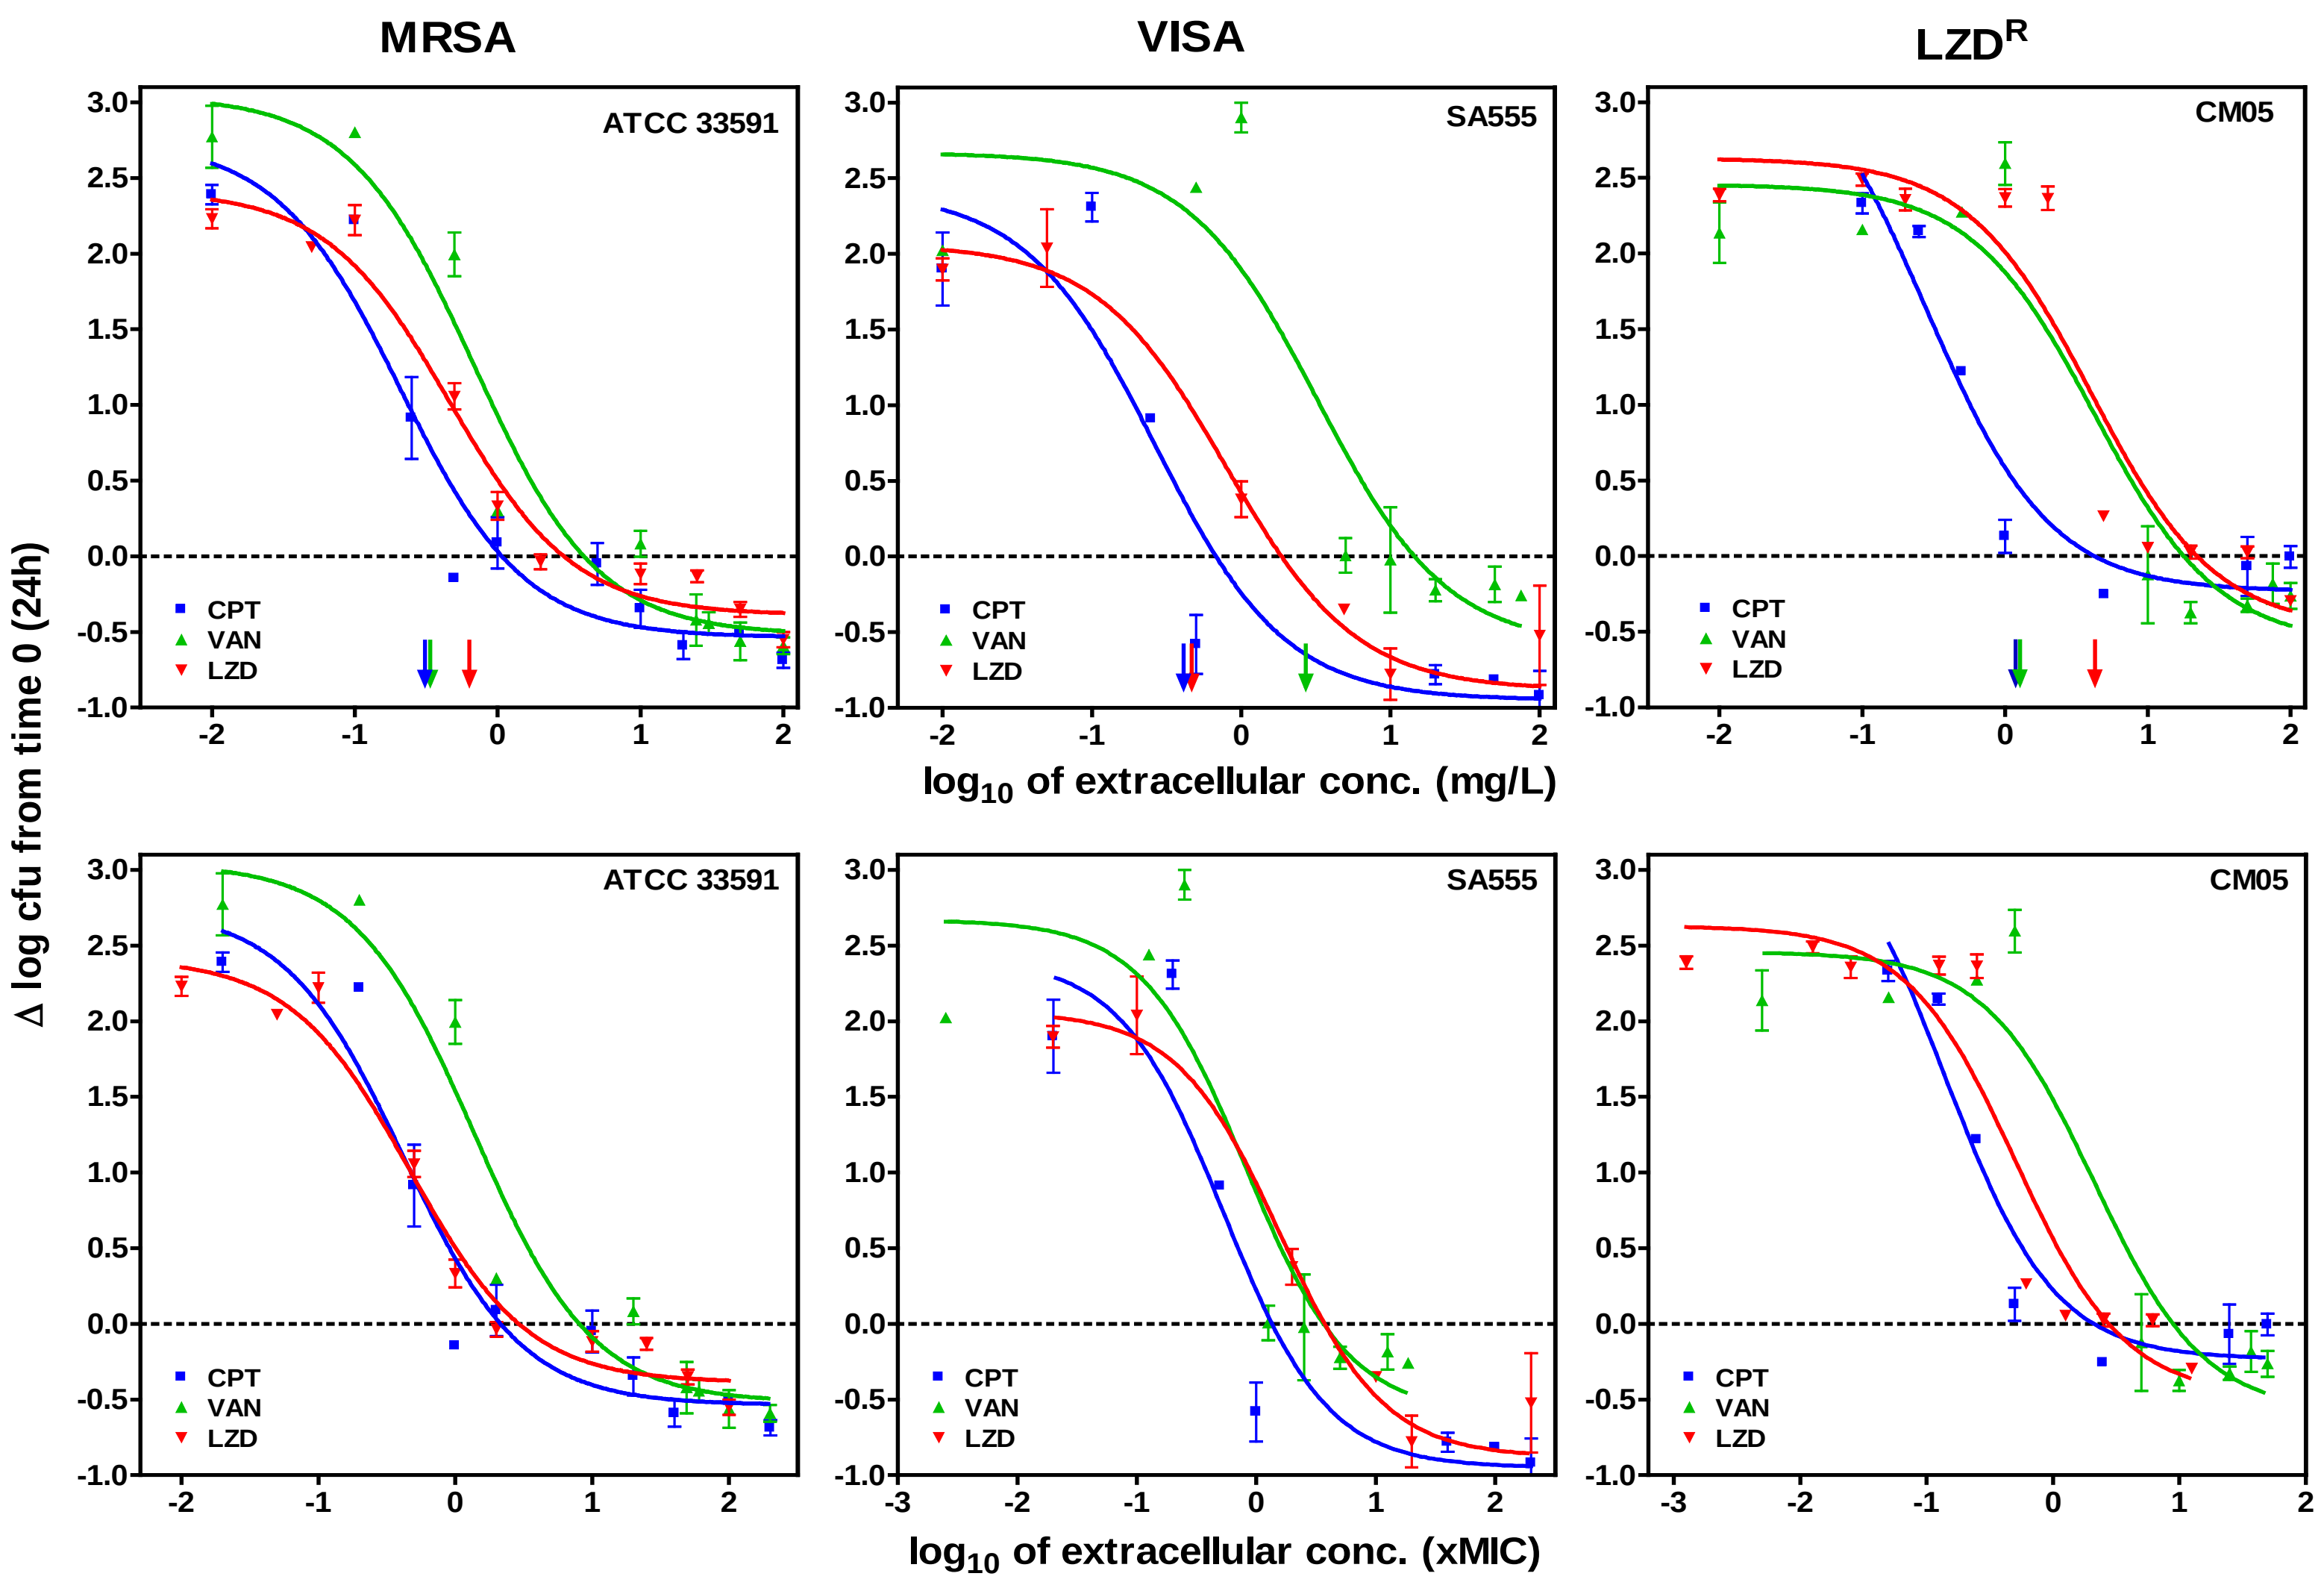
MRSA
VISA
LZD^R
ATCC 33591
SA555
CM05
CPT
VAN
LZD
Δ log cfu from time 0 (24h)
log10 of extracellular conc. (mg/L)
log10 of extracellular conc. (xMIC)

	CPT	VAN	LZD
MRSA (ATCC 33591)			
E_{max} (Δlog cfu)	-0.53 ± 0.16	-0.52 ± 0.19	-0.39 ± 0.11
EC_{50} (mg/L)	0.21	0.68	0.47
C_s (mg/L)	1.10	4.02	2.95
MRSA - VISA (SA 555)			
E_{max} (Δlog cfu)	-0.95 ± 0.36	-0.60 ± 0.52	-0.88 ± 0.17
EC_{50} (mg/L)	0.27	3.24	0.80
C_s (mg/L)	0.67	14.72	1.91
MRSA – LZD^R (CM05)			
E_{max} (Δlog cfu)	-0.23 ± 0.23	-0.58 ± 0.32	-0.48 ± 0.36
EC_{50} (mg/L)	0.28	4.24	4.03
C_s (mg/L)	4.17	17.71	22.91

Figure 1b. Intracellular Activity of CPT Towards Isolates Showing Increasing MIC Values to CPT

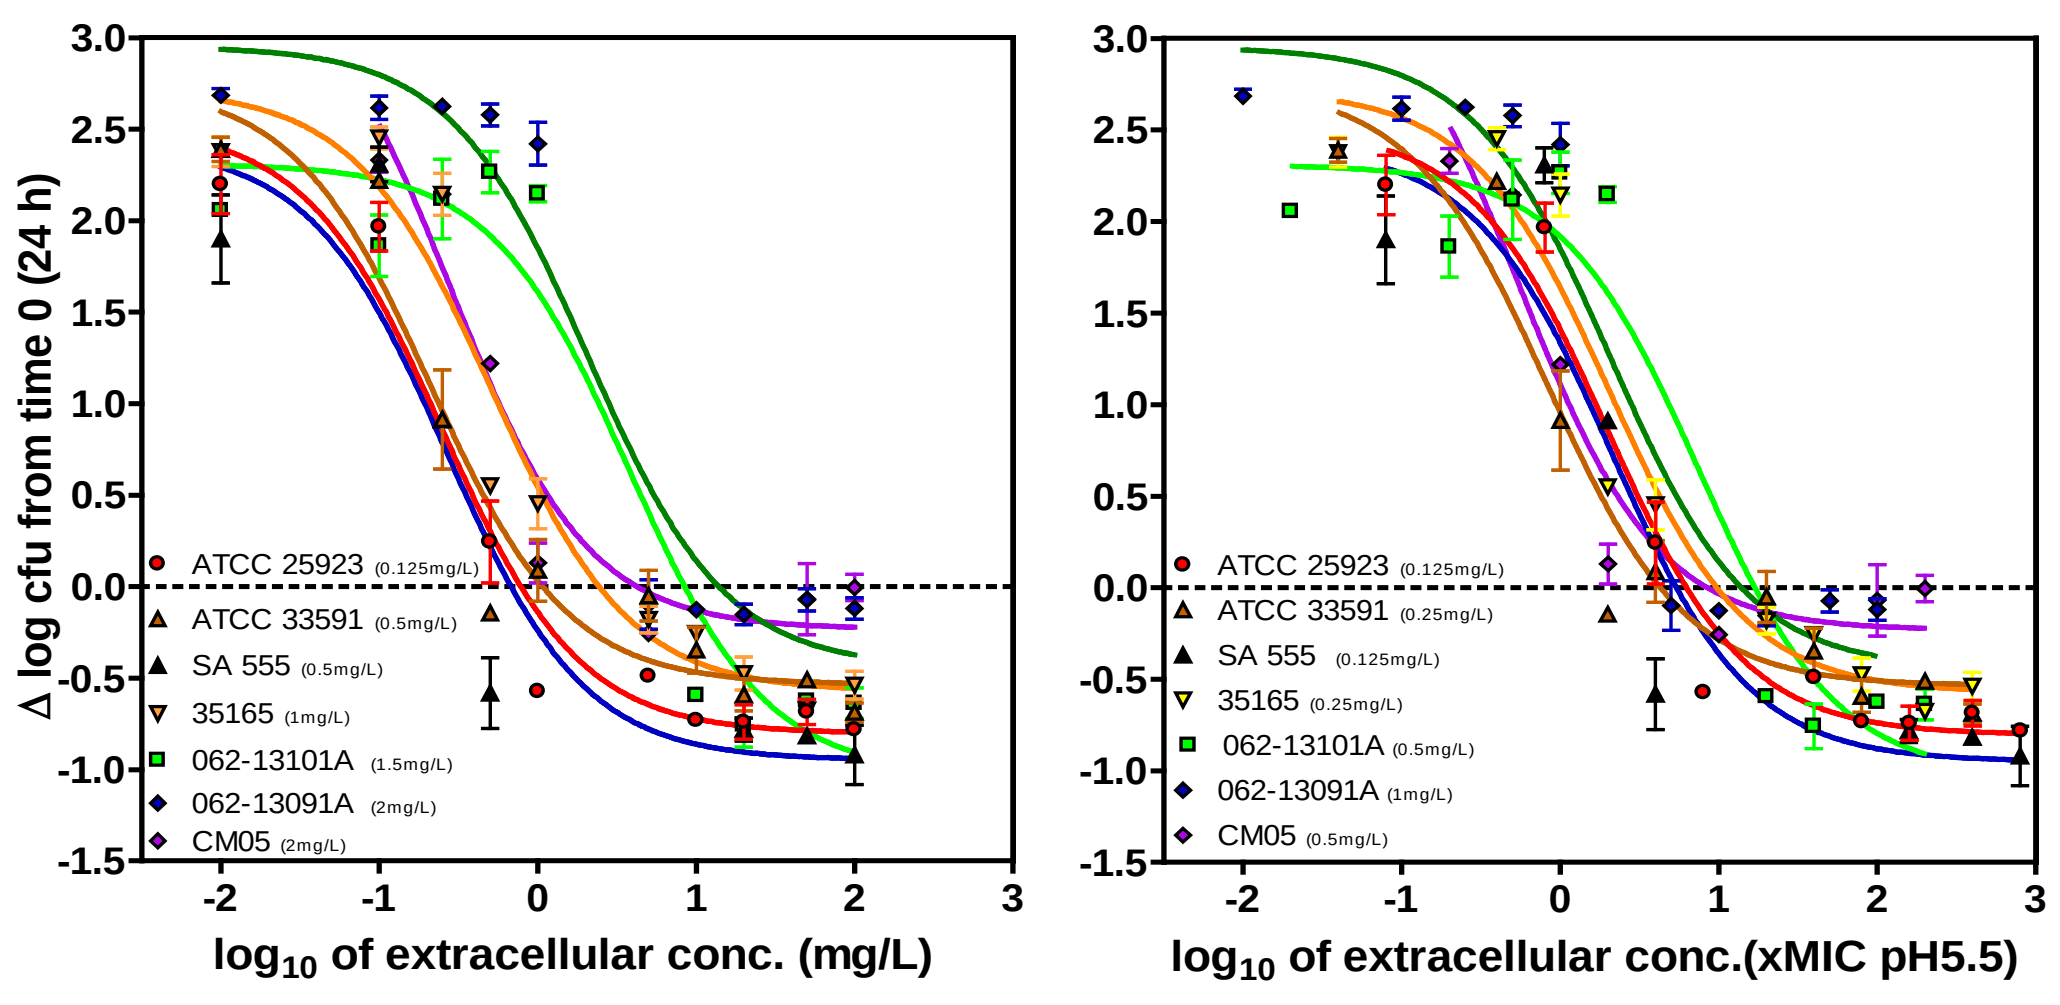
Δ log cfu from time 0 (24 h)
ATCC 25923 (0.125mg/L)
ATCC 33591 (0.5mg/L)
SA 555 (0.5mg/L)
35165 (1mg/L)
062-13101A (1.5mg/L)
062-13091A (2mg/L)
CM05 (2mg/L)
log10 of extracellular conc. (mg/L)
ATCC 25923 (0.125mg/L)
ATCC 33591 (0.25mg/L)
SA 555 (0.125mg/L)
35165 (0.25mg/L)
062-13101A (0.5mg/L)
062-13091A (1mg/L)
CM05 (0.5mg/L)
log10 of extracellular conc.(xMIC pH5.5)

Strain	E_{max} (Δlog cfu)	EC_{50} (mg/L)	C_s (mg/L)	C_s / MIC ratio for MIC at pH	
				7.4	5.5
ATCC 25923	-0.50 ± 0.13	0.25	0.79	6.3	6.3
ATCC 33591	-0.53 ± 0.16	0.21	1.10	2.2	4.4
SA 555	-0.95 ± 0.36	0.27	0.60	1.2	4.8
35165	-0.57 ± 0.17	0.51	1.40	1.4	5.6
062-13101A	-1.03 ± 0.36	3.76	8.50	5.7	17.0
062-13091A	-0.44 ± 0.27	2.08	13.80	6.9	13.8
CM05	-0.23 ± 0.23	0.28	4.17	2.1	8.3

Dose-response curves of antibiotics (top: CPT, VAN, LZD; bottom: CPT) against *S. aureus* with distinct resistance phenotypes after phagocytosis by THP-1 cells. Cells were incubated with the antibiotics for 24 h at the concentrations indicated on the abscissa (upper graphs: the vertical arrows point to the MIC of the antibiotics of the corresponding strains). The ordinate shows the change in the number of CFU per mg of cell protein as compared to the post-phagocytosis inoculum. All values are means of 2 to 3 determinations (± SD). The horizontal dotted line corresponds to an apparent static effect. Numeric data are shown in the Tables.

Conclusions

These *in vitro* data show that CPT is active against intraphagocytic *S. aureus* disregarding their resistance phenotype to VAN (and, to a lesser extent, to LZD) with essentially unimpaired values for E_{max} and C_s up to an MIC (at pH 7.4) of 2 and 1 mg/L, respectively. The higher C_s compared to MIC at pH 5.5 may be related to a poor intracellular accumulation, as observed with β-lactams in general.

This poster will be available for download after the meeting at http://www.facm.ucl.ac.be/posters.htm

References

1. Sinha and Fraunholz, *Int J Med Microbiol*. [2010] 300: 170-175.
2. Lemaire *et al* In: Methicillin-resistant *Staphylococcus aureus* (MRSA) – Etiology, at-risk populations and treatment, Nova Science Pub., New York [2009], pp. 55-74
3. Barcia-Macay *et al*, *Antimicrob Agents Chemother*.[2006] 50:841-51
4. Lemaire *et al*, *Antimicrob Agents Chemother*. [2009] 53: 2289-97.
5. Laudano JB, *J Antimicrob Chemother*. [2011] 66 Suppl 3: iii-11-18

Acknowledgments

We are grateful to M.C. Cambier for help with cell cultures. S.L. is postdoctoral fellow of the Belgian *Fonds de la Recherche Scientifique* (F.R.S.-FNRS). This work was supported by grant no. 3.597.06 of the Belgian *Fonds de la Recherche Scientifique Médicale* (F.R.S.M.) and a grant-in-aid from Cerexa Inc.

This study was supported in part by Cerexa, Inc. (a wholly-owned subsidiary of Forest Laboratories, Inc.). Cerexa, Inc. was involved in the design and decision to present these results. Cerexa, Inc. had no involvement in the collection, analysis, or interpretation of data. Scientific Therapeutics Information, Inc. provided editorial coordination and production services, which were funded by Forest Research Institute, Inc.