A-1291

Study of the Cellular Uptake and Subcellular Distribution of the Oxazolidinone Tedizolid (TZD) in Murine J774 Macrophages: Lack of Association with Mitochondria

Debaditya Das,[1] Axel Lambert,[1] Paul M.. Tulkens,[1] Giulio G. Muccioli,[2,3] Françoise Van Bambeke[1]

[1] Cellular and Molecular Pharmacology , [2] Mass Spectrometry Platform Analysis, [3] Bioanalysis and Pharmacology of Bioactive Lipids; Louvain Drug Research Institute, Université catholique de Louvain, Brussels, Belgium

<u>Mailing address</u>:
P.M. Tulkens
av. Mounier 73 Bte B1.73.05
1200 Brussels – Belgium,
tulkens@facm.ucl.ac.be

Abstract (edited)

Background: Linezolid (LZD) causes mitochondrial dysfunction resulting in potentially severe adverse effects (hyperlactacidemia, metabolic acidosis, and peripheral neuropathy). Tedizolid, (TZD, formerly known as torezolid, TR700 or DA7157), retains activity against LZD-resistant isolates. It accumulates about 10-fold more in macrophages compared to LZD, with commensurate larger activity against intraphagocytic *Staphylococcus aureus* (phagolysosomes), *Legionella pneumophila* (phagosomes), and *Listeria monocytogenes* (cytosol) [Lemaire et al., JAC, 2009; 64:1035-1043]. The large accumulation of tedizoid, however, raises questions about potential association with mitochondria.

Methods: Murine J774 macrophages were exposed to TZD (2-50 mg/L) for 2h, collected and homogenized for fractionation by differential (peletting) and isopycnic (sucrose gradient) centrifugation (Tulkens *et al.* J Cell Biol 1974; 63:383-401; Renard *et al.* AAC 1987; 31:410-6). TZD was quantified after extraction with $CHCl_3$:CH_3OH (8:4) by liquid chromatography (reverse phase) coupled with mass-spectrometry (LTQ-orbitrap; ESI; selective ion monitoring, with LZD as internal standard). TZD distribution was compared to that of marker enzymes for mitochondria (cytochrome *c*-oxidase [CYTOX]), lysosomes (N-acetyl-β-hexosaminidase [NaBGase]) and cytosol (lactate dehydrogenase [LDH]).

Results: After both differential and isopycnic cell fractionation, TDZ was consistently recovered in fractions corresponding to the cytosol and not detected in those enriched in mitochondria or lysosomes.

Conclusions: In spite of a higher accumulation in eukaryotic cells compared to LZD, TDZ seems not associated in a stable fashion to mitochondria. Observing TDZ in the cytosol after cell fractionation may point to its ability to diffuse throughout the cell, explaining it higher activity against organisms harbored in distinct subcellular compartments while at the same time not necessarily implying a higher potential mitochondrial toxicity.

References

1. Prokocimer *et al.* In Vitro Activity and Microbiological Efficacy of tedizolid (TR-700) against Gram-positive clinical isolates from a Phase 2 Study of oral tedizolid phosphate (TR-701) in patients with complicated skin and skin structure infections. Antimicrob Agents Chemother 2012 Jun 11. [Epub ahead of print]
2. Betriu *et al*. Comparative activities of TR-700 (torezolid) against staphylococcal blood isolates collected in Spain. Antimicrob Agents Chemother. 2010 May; 54(5):2212-5.
3. Lemaire *et al.* Cellular pharmacokinetics and intracellular activity of torezolid (TR-700): studies with human macrophage (THP-1) and endothelial (HUVEC) cell lines. J Antimicrob Chemother. 2009 Nov; 64(5):1035-43.
4. Housman *et al.* Pulmonary disposition of tedizolid following administration of once-daily oral 200-milligram tedizolid phosphate in healthy adult volunteers. Antimicrob Agents Chemother. 2012 May; 56(5):2627-34.
5. Garrabou *et al.* Reversible inhibition of mitochondrial protein synthesis during linezolid-related hyperlactatemia. Antimicrob Agents Chemother, 51 (2007), pp. 962–967.
6. McKee *et al.* Inhibition of mammalian mitochondrial protein synthesis by oxazolidinones. Antimicrob Agents Chemother. 2006 Jun; 50(6):2042-9.
7. Leach *et al.* The site of action of oxazolidinone antibiotics in living bacteria and in human mitochondria. Mol Cell. 2007 May 11; 26(3):393-402.
8. Lemaire *et al.* Cellular pharmacokinetics of the novel biaryloxazolidinone radezolid in phagocytic cells: studies with macrophages and polymorphonuclear neutrophils. Antimicrob Agents Chemother. 2010, Jun; 54(6):2540-8.
9. de Duve *et al.* Lysosomotropic agents. Biochem Pharmacol. 1974 Sep 15; 23(18):2495-531.
10. de Duve C. Exploring cells with a centrifuge. Nobel Lectures, Physiology or Medicine 1971-1980, Editor Jan Lindsten, World Scientific Publishing Co., Singapore, 1992 – available on line at http://www.nobelprize.org/nobel_prizes/medicine/laureates/1974/duve-lecture.pdf
11. Tulkens PM. Intracellular distribution and activity of antibiotics. Eur J Clin Microbiol Infect Dis. 1991 Feb; 10(2):100-6.

Background and Aim

There is an continuous need of discovering and developing new antibiotics to treat infections caused by multidrug-resistant pathogens. In this context, new oxazolidinones have emerged such as tedizolid (also referred to as torezolid or TR-700) that show higher intrinsic activities (lower MICs) than linezolid against Gram-positive cocci from clinical origin, including MRSA, and unimpaired activity against *cfr*+ linezolid-resistant strains [1,2].

Tedizolid also accumulates about 10-fold more than linezolid in human THP-1 macrophages *in vitro* [3] and accumulates about 20-fold over serum concentrations in alveolar macrophages of healthy adult volunteers [4]. While this may confer an advantage in terms of activity against intracellular forms of target bacteria [3], it also raises concerns with respect to potential toxicities. Indeed, one of the main likely mechanisms for the long-term toxicities of linezolid probably relates to its ability to impair mitochondrial DNA-dependent protein synthesis [5,6], due to structural similarities between mitochondrial and procaryotic ribosomes [7].

Surprisingly, however, there are no published studies examining the subcellular localization of linezolid. Conversely, we know that another advanced oxazolidinone, radezolid, which also accumulates more in human THP-1 cells than linezolid, is mainly localized in lysosomes [8]. This can be rationalized based on the presence of aminofunction with a pKa ~7.4 (see structure) resulting in a proton-trapping of its ionized form in acidic membrane-bounded organelles [9]. Tedizolid, however, has no such aminofunction and remains essentially uncharged at pHs 2 to 11, making prediction of its subcellular localization difficult.

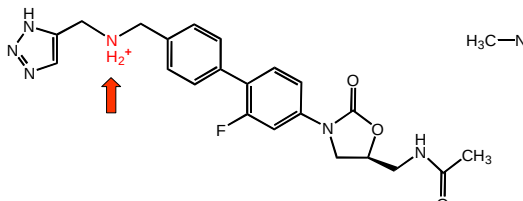

radezolid

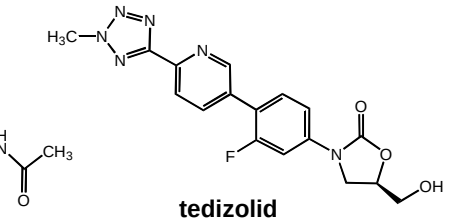

tedizolid

The present study, therefore, is aimed at studying the subcellular distribution of tedizolid.

Methods

We used the method of cell fractionation, consisting in collecting cells incubated with the drug of interest, disrupting their pericellular membrane without damaging the integrity of the subcellular organelles (homogenization) and separating these organelles by subjecting the suspension to centrifugation at increasing speeds (differential pelletting, in which organelles are mainly separated based on their size) or by centrifugation through sucrose gradients (isopycnic equilibration) in which organelles are mainly separated based on their buoyant density). Fractions are thereafter assayed for the presence of markers of the organelles of interest and for drug content.

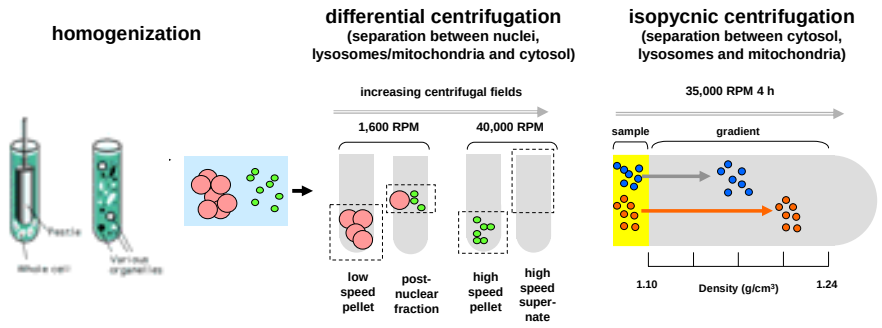

homogenization
differential centrifugation
(separation between nuclei, lysosomes/mitochondria and cytosol)
isopycnic centrifugation
(separation between cytosol, lysosomes and mitochondria)
increasing centrifugal fields
1,600 RPM
40,000 RPM
35,000 RPM 4 h
sample
gradient
low speed pellet
post-nuclear fraction
high speed pellet
high speed super-nate
1.10
Density (g/cm³)
1.24

These methods, originally developed to discover and characterize subcellular organelles [10], have been previously used and validated for the study of the subcellular localization of aminoglycosides (lysosomes), macrolides (2/3 in lysosomes; 1/3 in cytosol) and fluoroquinolones (cytosol) [11].

Murine J774 macrophages were selected because these can easily be fractionated and mitochondria satisfactorily separated from lysosomes by isopycnic centrifugation.

Cells were incubated for 2 h with 20 mg/L tedizolid before being harvested and subjected to fractionation.

Marker enzymes were:

- for lysosomes: N-acetyl-β-hexosaminidase (NABgase)
- for mitochondria: cytochrome *c*-oxidase (cyt-oxidase)
- for cytosol: lactate-dehydrogenase (LDHase)

Analytical method:

Tedizolid was extracted from cell fractions by phase partition and protein precipitation in chloroform:methanol (8:4), recovered in methanol and quantified by liquid chromatography (reverse phase) coupled to mass spectrometry (LTQ-orbitrap). The gradient (0.4 mL/min) was designed as follows : from methanol-water (1:1) to methanol in 20 min, then 5 min with methanol, and subsequent equilibration in the initial conditions. (typical tedizolid elution time: 7 min). Acetic acid (0.5%) was used to assist ionization in the positive mode. Linezolid was used as an internal standard, with linear response obtained in a 10-1,000 ng/mL range.

Results

1. Differential centrifugation

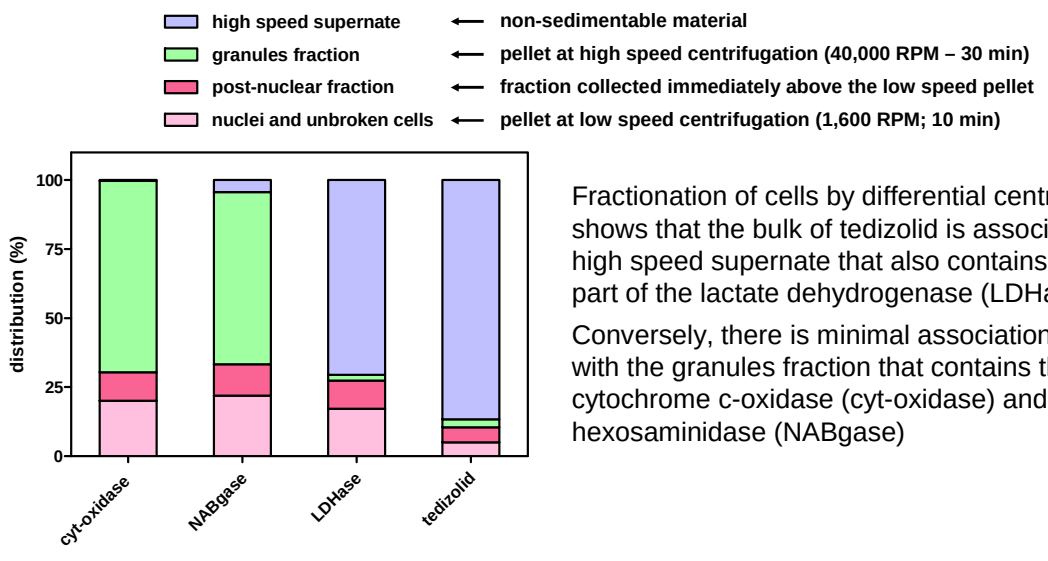

high speed supernate ← non-sedimentable material
granules fraction ← pellet at high speed centrifugation (40,000 RPM – 30 min)
post-nuclear fraction ← fraction collected immediately above the low speed pellet
nuclei and unbroken cells ← pellet at low speed centrifugation (1,600 RPM; 10 min)
distribution (%)
100
75
50
25
0
cyt-oxidase
NABgase
LDHase
tedizolid

Fractionation of cells by differential centrifugation shows that the bulk of tedizolid is associated with the high speed supernate that also contains the largest part of the lactate dehydrogenase (LDHase).

Conversely, there is minimal association of tedizolid with the granules fraction that contains the bulk of cytochrome c-oxidase (cyt-oxidase) and N-acetyl-β-hexosaminidase (NABgase)

2. Isopycnic centrifugation (after elimination of nuclei and unbroken cells)

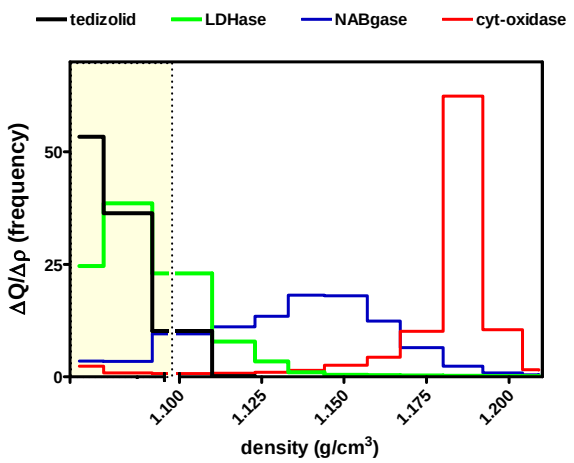

tedizolid
LDHase
NABgase
cyt-oxidase
ΔQ/Δρ (frequency)
50
25
0
1.100
1.125
1.150
1.175
1.200
density (g/cm³)

After isopycnic centrifugation, the bulk of tedizolid is recovered in the top fractions corresponding to where material was deposited (zone highlighted in light yellow), indicating no association with organelles that have moved into the gradient and have equilibrated at their buoyant density. The separation from mitochondria (cyt-oxidase) is strikingly complete. There is also no sign of association with lysosomes (NABgase). The slight migration of LDHase is due to the size of this protein that allows for migration due to the high centrifugation field used.

Conclusions

1. There is no evidence of any stable association of tedizolid with mitochondria of eukaryotic cells in spite of its large cellular accumulation (compared to linezolid). Tedizolid, rather, seems to be free in cytosol.
2. While we cannot exclude post-homogenization desorption of tedizolid from an intracellular storage site, the data rather suggest that the intracellular drug may be essentially free, which may explain its enhanced activity against intracellular bacteria but does not exclude a reversible inhibitory activity against mitochodrial protein synthesis.
3. Additional studies with longer incubation times, measurement of mitochondrial protein synthesis, and comparison with linezolid and other oxazolidinones may help in better defining the significance of the present data.

Acknowledgements

We thank M.C. Cambier for help with cell cultures and M. Vergauwen for excellent and constant technical assistance.

This work was supported by the Belgian *Fonds de la Recherche Scientifique Médicale* (*FRSM*) and by a grant-in-aid from Trius Therapeutics.

This poster will be available for download after the meeting from http://www.facm.ucl.ac.be/posters.htm