Poster A1-1936
49th ICAAC
San Francisco, CA, USA
September 12-15, 2009

Accumulation and Intracellular Activity of Radezolid, a New Oxazolidinone, in non-Phagocytic Cells

S. Lemaire[1], K. Kosowska-Shick[2], P. C. Appelbaum[2], J.-P. Pirnay[3], G. Verween[3], P. De Corte[3], D. De Vos[3], P. M. Tulkens[1] and F. Van Bambeke[1]

[1] Université catholique de Louvain, Brussels, Belgium; [2] Hershey Medical Center, Hershey, PA, USA; [3] Queen Astrid Military Hospital, Brussels, Belgium

Mailing address:
F. Van Bambeke
UCL 73.70 av. Mounier 73,
1200 Brussels,
Belgium
francoise.vanbambeke@uclouvain.be

ABSTRACT

Background. *S. aureus* invades and persists within a variety of non-phagocytic cells. This may play a determining role in relapsing diseases like skin or respiratory tract infections, osteomyelitis, and endocarditis. Effective treatment may require drug accumulation in target cells. We have previously shown that radezolid (RDZ) accumulates 8-fold in human macrophages (Lemaire et al. ECCMID 2009, O29). We have now determined its intracellular accumulation and activity in a series of relevant non-phagocytic cells.

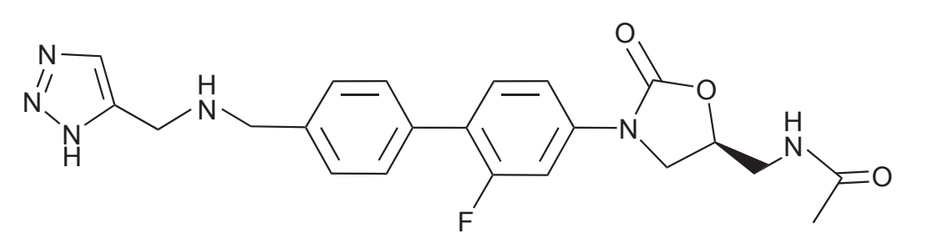

Radezolid

Figure 1

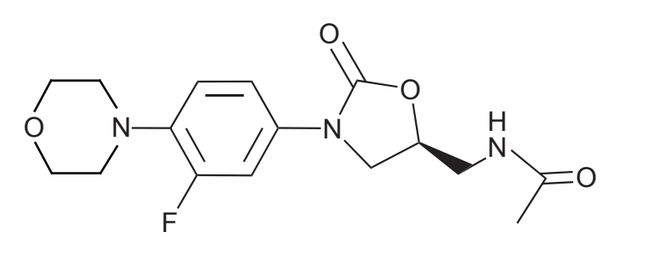

Linezolid

Methods. Cells are listed in the table. Cellular concentration was measured at 24h using ^{14}C-RDZ, and accumulation calculated using a mean cell volume of 5 µl/mg cell prot. MICs and dose-effect relationships at 24h were determined as described (Barcia-Macay et al, AAC, 2006), using MRSA SA238 (clinical isolate) and a linezolid (LZD)^R *in vitro* mutant thereof (SA238L). Results are expressed as the change in 24h inoculum compared to t=0.

Results. The cellular accumulation of RDZ and its intracellular activity (against *S. aureus* strain SA238, [MICs: 0.5-1 and 2 mg/L for RDZ and LZD] was similar in all cell types. Against the LZD^R mutant (MICs: 2 and 8 mg/L for RDZ and LZD), RDZ C_{static} and E_{max} remained constant while LZD activity was diminished (C_{static} range from 15.7 mg/L in Calu-3 to > 100 mg/L in osteoblasts; E_{max}, –0.6 log in keratinocytes to +0.5 log in osteoblasts).

Conclusions. RDZ accumulates in non-phagocytic cells to the same extent as in macrophages and shows comparable intracellular activity against both LZD^S and LZD^R MRSA. LZD was systematically less effective against the resistant strain.

BACKGROUND AND AIM

It is now well accepted that *Staphylococcus aureus* is a facultative intracellular pathogen and that its capacity to survive within non-phagocytic cells may play an important role in the persistent and recurrent character of several infections, such as complicated skin and soft tissue infections, osteomyelitis, endocarditis, or pulmonary infections associated with cystic fibrosis (1).
Effective treatment of such infections may therefore require the use of antibiotics accumulating intracellularly and demonstrating activity therein.
In this context, we have shown that radezolid (RDZ), a novel oxazolidinone antibiotic (see structure in Figure 1, and description on poster A1-1937), markedly accumulates (approx. 10-fold) within phagocytic cells (PMN and macrophages) and shows improved potency towards both linezolid-susceptible and linezolid-resistant *S. aureus* phagocytized by human THP-1 macrophages (2; A1-1937).
The aim of the present study was to measure the intracellular accumulation and activity of radezolid in a series of non-phagocytic cells originating from the tissues subject to persistent infections.

RESULTS

Cellular accumulation of radezolid

Cell lines	cellular accumulation (24h)
Rat fibroblasts (primary culture)	10.9 ± 2.1
Human skin keratinocytes (primary cultures)	16.1 ± 2.0
Normal human osteoblasts (NHost)	9.7 ± 0.4
Human Umbilical Vein endothelial cells (HUVEC)	10.5 ± 0.1
Human lung epithelial cells (Calu-3)	8.3 ± 1.0

At 24 h, radezolid accumulates to a high and similar level within all non-phagocytic cells, reaching an accumulation factor similar to that observed within phagocytic cells (2).

Intracellular activity of antibiotics

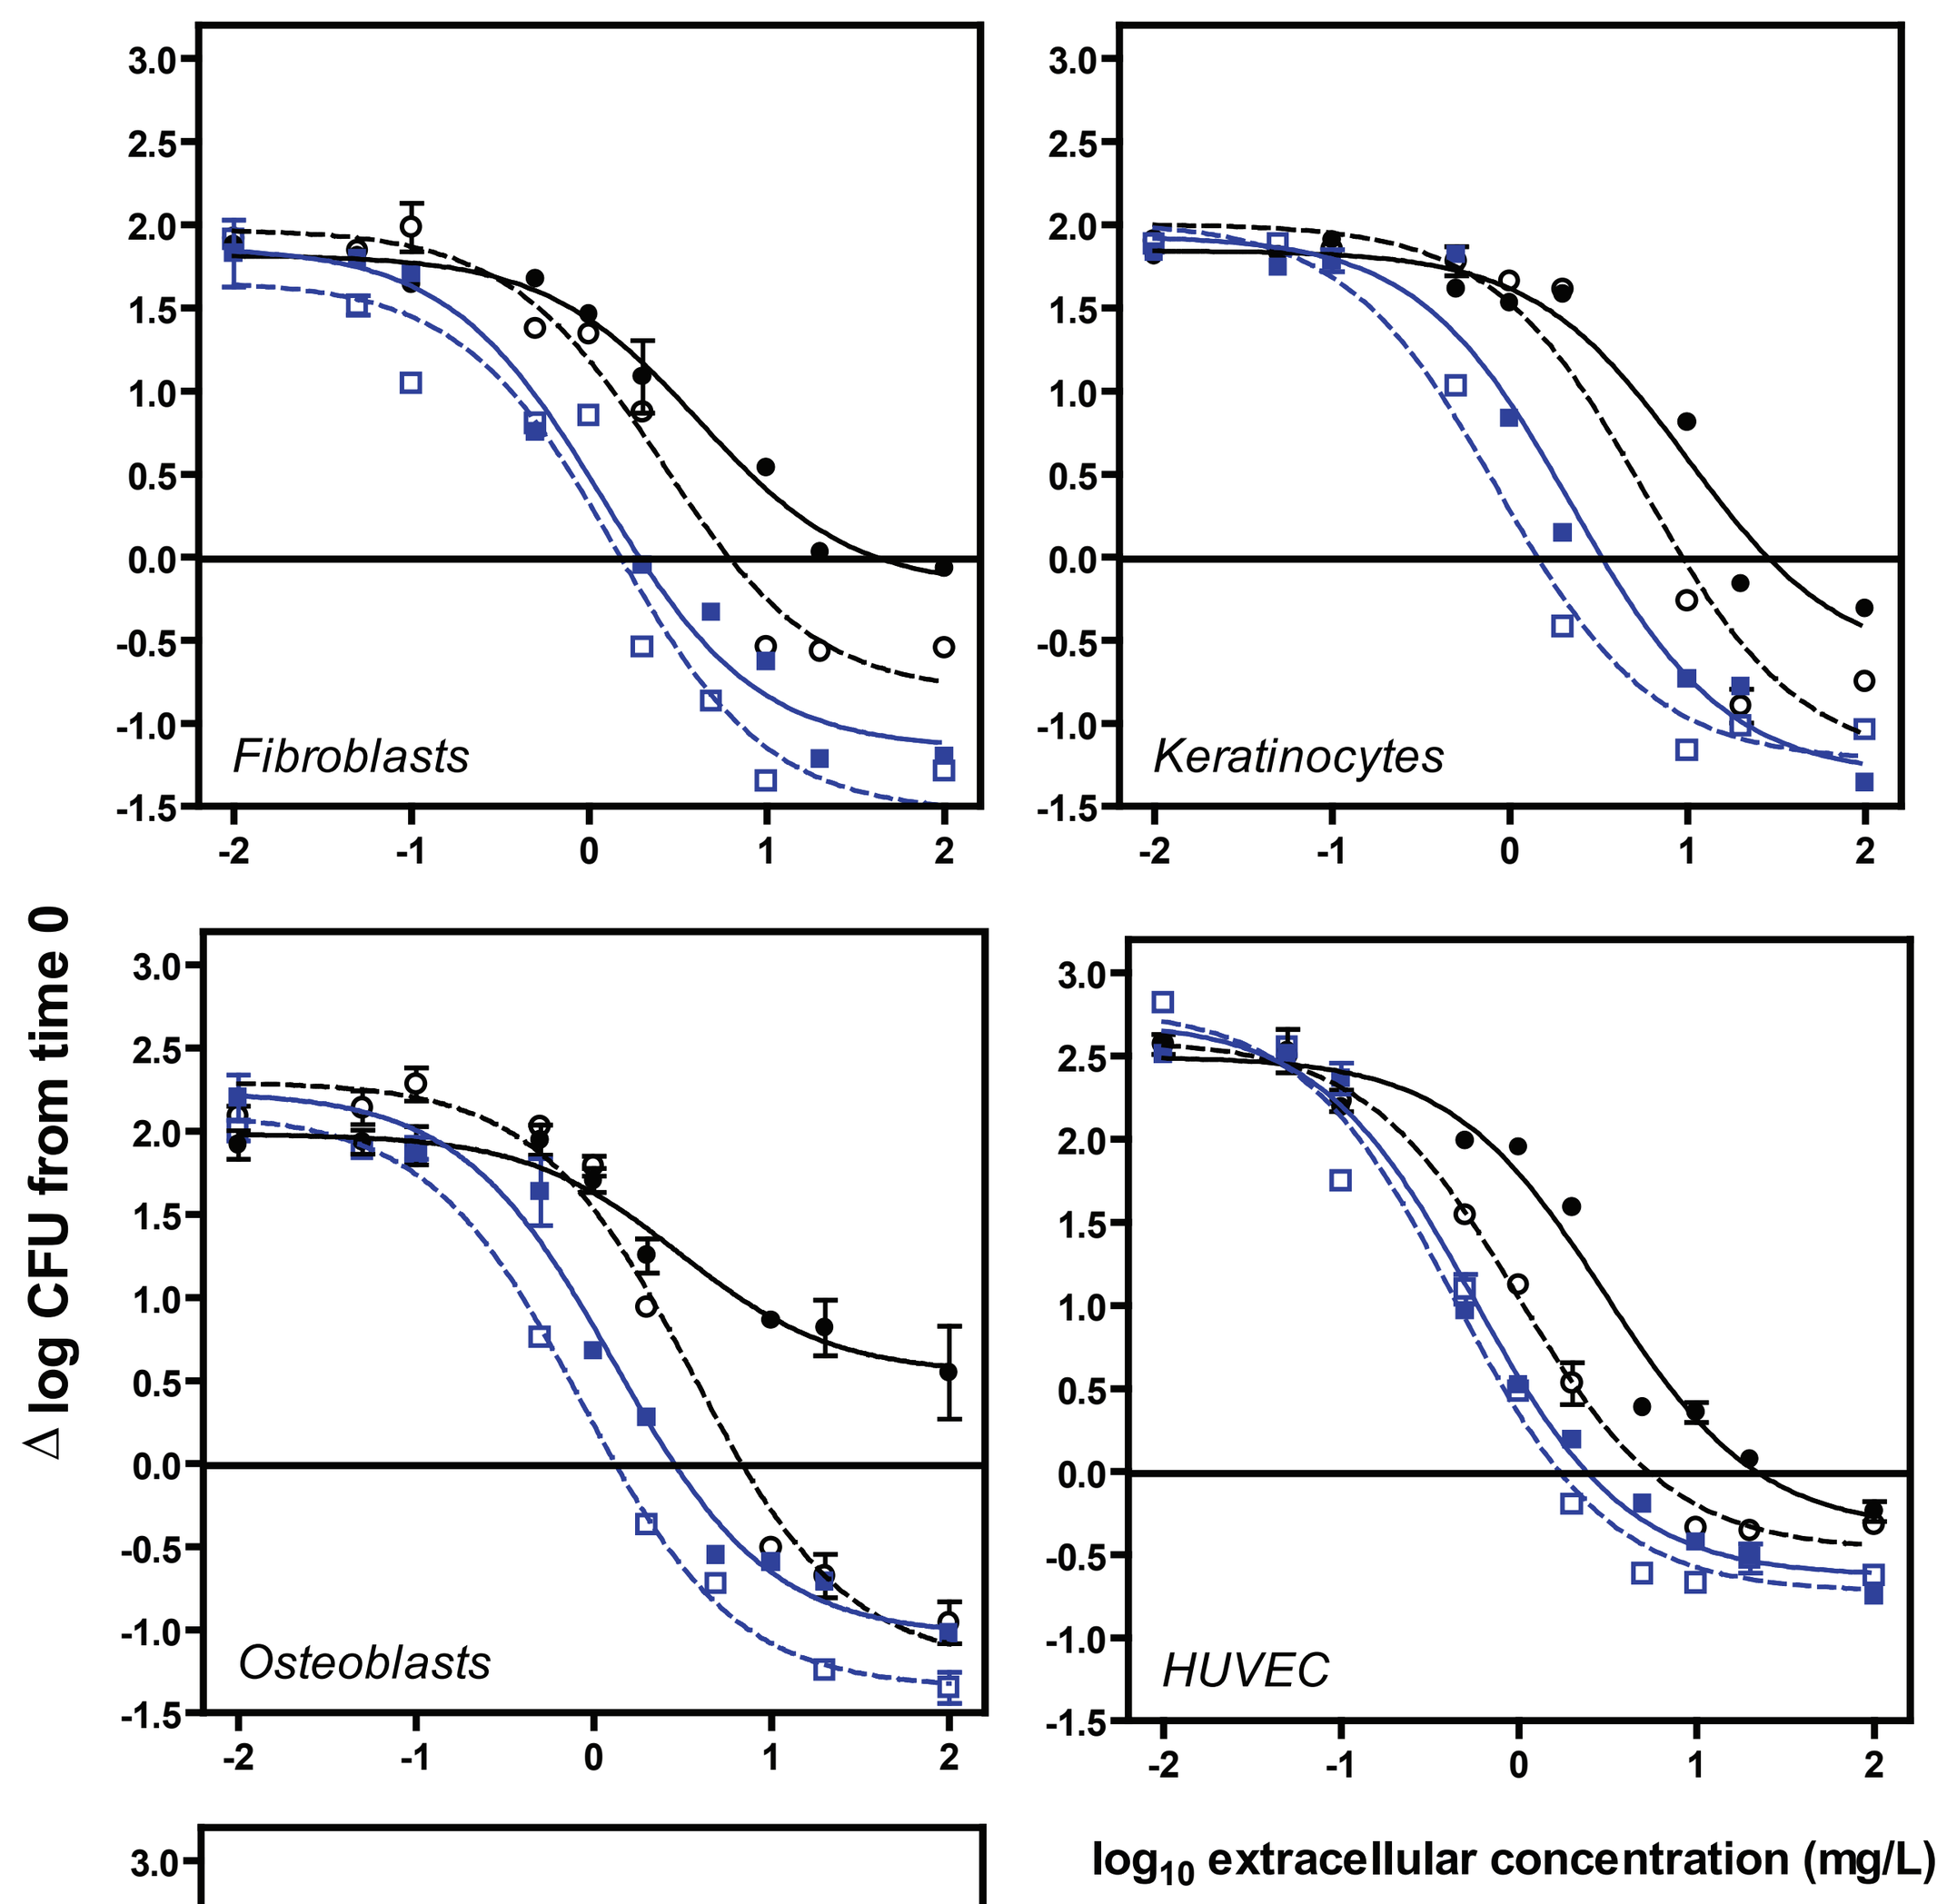
Fibroblasts
Keratinocytes
Osteoblasts
HUVEC
Δ log CFU from time 0
log10 extracellular concentration (mg/L)

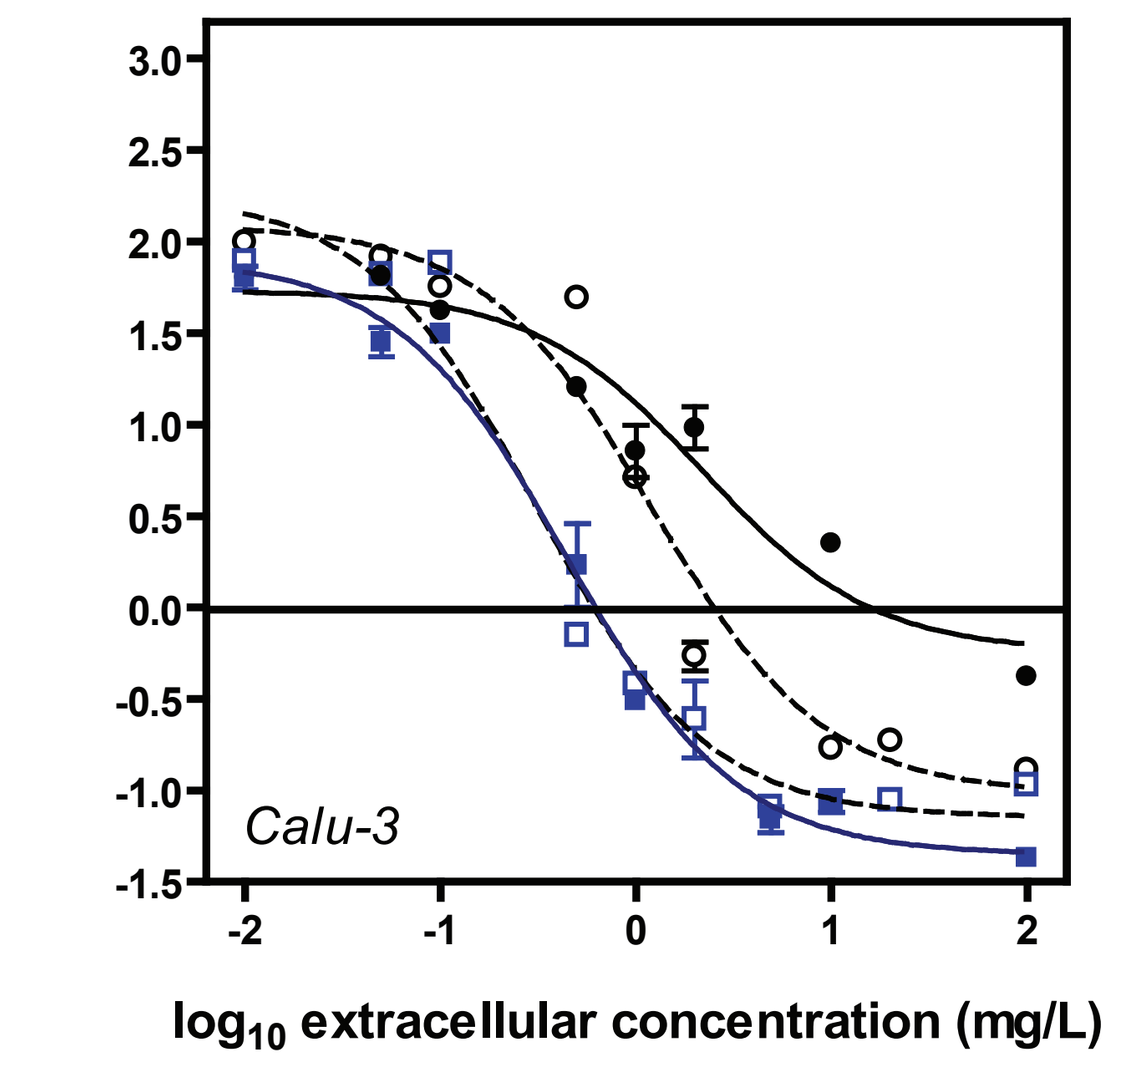
Calu-3
log10 extracellular concentration (mg/L)

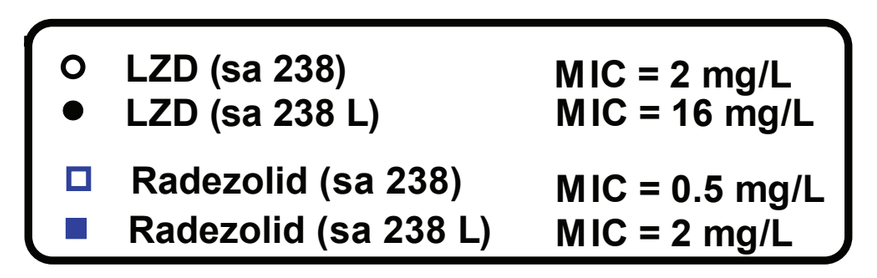
LZD (sa 238) MIC = 2 mg/L
LZD (sa 238 L) MIC = 16 mg/L
Radezolid (sa 238) MIC = 0.5 mg/L
Radezolid (sa 238 L) MIC = 2 mg/L

Figure 2. Concentration-killing effect of linezolid vs. radezolid against linezolid-susceptible SA238 and linezolid-resistant (SA238L) strains phagocytosed by different cell types. Cells were incubated with antibiotics for 24 h in the presence of increasing concentrations of antibiotics. The graph shows the change in CFU per mg of cell protein.

Pharmacological descriptors as calculated by non-linear (sigmoid) regresion of the dose-response curves illustrated in Figure 2.

strain	Cell line	linezolid		radezolid		Cstatic (LZD/RDZ ratio)
		C_{static}[a] (mg/L)	E_{max}[b]	C_{static}[a] (mg/L)	E_{max}[b]	
SA238 (LZD-S)	Fibroblasts	~ 6.6	-0.8 ± 0.1	**~ 1.5**	**-1.5 ± 0.3**	4.4
	Keratinocytes	~ 9.5	-1.2 ± 0.3	**~ 1.5**	**-1.2 ± 0.1**	6.3
	Osteoblasts	~ 6.8	-1.2 ± 0.2	**~ 1.3**	**-1.3 ± 0.1**	5.2
	HUVEC	~ 5.8	-0.5 ± 0.1	**~ 1.7**	**-0.7 ± 0.1**	3.4
	Calu-3	~ 2.6	-1.2 ± 0.2	**~ 0.6**	**-1.3 ± 0.1**	4.3
SA238L (LZD-R)	Fibroblasts	~ 42	-0.2 ± 0.1	**~ 1.9**	**-1.2 ± 0.1**	22
	Keratinocytes	~ 27	-0.6 ± 0.3	**~ 3.3**	**-1.3 ± 0.2**	8.2
	Osteoblasts	> 100	+0.5 ± 0.1	**~ 2.8**	**-1.0 ± 0.1**	> 35
	HUVEC	~ 20	-0.4 ± 0.1	**~ 2.3**	**-0.6 ± 0.1**	8.7
	Calu-3	~ 15	-0.2 ± 0.2	**~ 0.6**	**-1.4 ± 0.1**	25

[a] Static concentration : concentration yielding no apparent change in the inoculum compared to time 0
[b] Maximal Efficacy: Change in log10 CFU compared to time 0 for an infinitely large concentration of antibiotic

- Radezolid shows similar efficacy (E_{max}) and potency (C_{static}) against both the SA238 susceptible and the SA238L resistant strains.
- Radezolid is 3.4- to 6.3-fold more potent than linezolid against the SA238 susceptible strain and 8.7 to > 100 fold against the SA238L resistant strain.
- Although MICs are the same (2 mg/L) , radezolid shows 2-3-fold higher potency against the resistant strain SA238L than linezolid against the susceptible strain SA238.
- Radezolid shows similar efficacy in all cell types; linezolid efficacy is reduced against the SA238L resistant strain in all cells.

METHODS

Bacteria and susceptibility testings. *S. aureus* SA238 and SA238L, a linezolid-resistant mutant thereof selected in vitro, were used in the present study (3). MICs were determined in Mueller Hinton Broth as previously described (4).

Cell lines. Experiments were conducted with primary cultures of rat fibroblasts or of human skin keratinocytes, and with continous cell lines of human lung epithelial cells (ATCC, Manassas, VA), normal human osteoblasts (Lonza), and human umbilical vein endothelial cells (HUVEC, Lonza).

Determination of the cellular accumulation. The cellular content of radezolid was determined by scintillation, cells having been incubated with ^{14}C-RDZ.

Cell infection and assessment of the intracellular activities of antibiotics. Infection of cells was performed exactly as described before (3-5) with typical starting inoculum of ~ 1 to 2 x10^6 CFU/mg prot. Antibiotic activity was examined after 24 h in the presence of increasing concentrations of antibiotics (0.01- to 100 mg/L), and results were expressed as the change in the inoculum at 24 h compared to time 0 (4-5). Data were analyzed by non-linear regression using Hill's equation to calculate pharmacological descriptors (E_{max}, maximal reduction of the intracellular inoculum [in log_{10} units] for an infinitely large antibiotic concentration; EC_{50} , antibiotic concentration [in mg/L] yielding a response half-way between E_0 and E_{max}, as obtained from the Hill equation), together with the static concentration (C_{static}) estimated by graphical interpolation.

This poster will be made available for download after the meeting:
http://www.facm.ucl.ac.be/posters.htm

We thank M.C. Cambier and C. Misson for technical assistance. S.L. and F.V.B. are Post-doctoral and Senior Research Associate of the Belgian Fonds de la Recherche Scientifique (FSR-FNRS). This work was supported by the Belgian Fonds de la Recherche Scientifique Médicale (FRSM) and a grant-in-aid from Rib-X Pharmaceuticals Inc.

CONCLUSIONS

- **Radezolid accumulates to a similar extent in all cell types.**
- **Radezolid shows globally a similar efficacy (E_{max}) relative to linezolid, but a higher relative potency (lower C_{static}) than linezolid against intracellular *S. aureus* in all cell types.**
- **Radezolid shows similar intracellular activities (E_{max} and C_{static}) against linezolid-susceptible and linezolid resistant strains, suggesting that it could be a useful substitute to linezolid to treat intracellular infections caused by these resistant bacteria.**

REFERENCES

(1) Garzoni & Kelley. Trends Microbiol. (2009) 17:59-65.
(2) Lemaire et al. 26th ICC, Toronto, ON (2009)
(3) Kosowska-Shick et al. Antimicrob. Agents Chemother. (2006) 50:765-769.
(4) Barcia-Macay et al. Antimicrob. Agents Chemother. (2006) 50: 841-851.
(5) Lemaire et al, Antimicrob. Agents Chemother. (2008) 52: 2797-2805.