Comparative activity of moxifloxacin vs. trimethoprim-sulfamethoxazole, cloxacillin, linezolid, clindamycin, and ciprofloxacin against intracellular methicillin-sensitive (MSSA) and community-acquired methicillin-resistant (CA-MRSA) *S. aureus*

S. Lemaire,[a] F. Van Bambeke,[a] P.M. Tulkens,[a] and Y. Glupczynski[b]

[a]Unité de pharmacologie cellulaire et moléculaire, Université catholique de Louvain, Brussels;
[b]Laboratoire de microbiologie, Cliniques universitaires UCL Mont-Godinne, Yvoir; Belgium

Mailing address:

Sandrine Lemaire
Pharmacologie cellulaire et moléculaire
UCL 73.70 av. Mounier 73
1200 Brussels - Belgium
Sandrine.lemaire@facm.ucl.ac.be

P 703

Abstract

Objectives: Recurrence and persistence of *S. aureus* infection is often ascribed to intracellular bacterial persistence and ensuing emergence of resistance. We show that the activity of antibiotics may markedly differ when tested against extracellular and intracellular bacteria (AAC 2006, 50:841-851). In this context, we have examined the intraphagocytic activity of moxifloxacin (MXF, recently approved for skin and soft-tissues infections) in comparison to (i) antibiotics commonly used for the treatment of CA-MRSA infections (trimethoprim-sulfamethoxazole [TMP-SMX], cloxacillin [CLX], linezolid [LNZ], clindamycin [CLI], and (ii) ciprofloxacin [CIP] against fully-sensitive *S. aureus* (MSSA) and the newly-emerging commynity-acquired methicillin-resistant *S. aureus* (CA-MRSA).

Methods: MSSA (ATCC 25923) AND CA-MRSA (NRS 192) were used. MICs were determined by micro-dilution in MH broth. Intracellular activity was assessed on phagocytosed MSSA or CA-MRSA by THP-1 macrophages (as described in details in JAC 2005, 55:897-904) after 24 h exposure to drug concentrations corresponding to the respective human Cmax (see Table). Controls cells (no antibiotics added) were incubated with gentamicin [0.5 x MIC] to prevent extracellular growth (validation in AAC 2006 50:841-851).

Results: The table shows the MICs of each drug, together with the corresponding intracellular activity (change in cfu from the post-phagocytosis inoculum).

Drugs (Cmax)	ATCC 25923 (MSSA)		NRS 192 (CA-MRSA)	
	MIC (mg/L)	Intr. Act. (Δ log cfu)	MIC (mg/L)	Intr. Act. (Δ log cfu)
TMP-SMX (25)	1	+0.6 ± 0.1	1-2	+0.7 ± 0.1
CLX (8)	0.125	-0.8 ± 0.1	0.5-1	-0.7 ± 0.1
LNZ (21)	1	-0.7 ± 0.1	2	-0.7 ± 0.1
CLI (12)	0.06	-1.0 ± 0.1	0.125	-0.9 ± 0.1
CIP (4)	0.125	-1.3 ± 0.1	0.5	-1.4 ± 0.1
MXF (4)	0.03	-2.0 ± 0.0	0.03	-1.8 ± 0.1

Conclusions : All antibiotics tested, with the exception of TMP-SMX, reduced the intracellular inoculum, but MXF displayed the largest effect (~ 2 log decrease), probably in relation with its low MIC towards the strains examined.

Background

Selecting an optimal treatment against *S. aureus* infections is facing two major issues, namely
(i) the increasing emergence of resistance to first line antibiotics, including in the community;
(ii) the difficulty of eradicating intracellular forms.[1]

In this context, moxifloxacin (MXF) appeared to us of interest because of (i) its bactericidal activity against intracellular MSSA as shown in an *in vitro* model of infected human THP-1 macrophages,[2] and (ii) its inhibitory effect on the expression of leucotoxins and virulence regulatory factors of *S. aureus*.[3]

We have therefore assessed the intracellular activity of MXF in comparison with ciprofloxacin (CIP) and other antibiotics commonly used for the treatment of CA-MRSA infections.

Results

1 Susceptibility testing

	Abbrev.	MICs (mg/L) MSSA[a]	CA-MRSA[b]
Trimethoprim-Sulfamethoxazole	TMP-SMX	1	1-2
Cloxacillin	CLX	0.125	0.5-1
Linezolid	LNZ	1	2
Clindamycin	CLI	0.06	0.125
Ciprofloxacin	CIP	0.125	0.5
Moxifloxacin	MXF	0.03	0.03

[a] ATCC 25923
[b] NRS 192 (Ery: 1 mg/L; *mecA* +)

2 Comparative intracellular activity

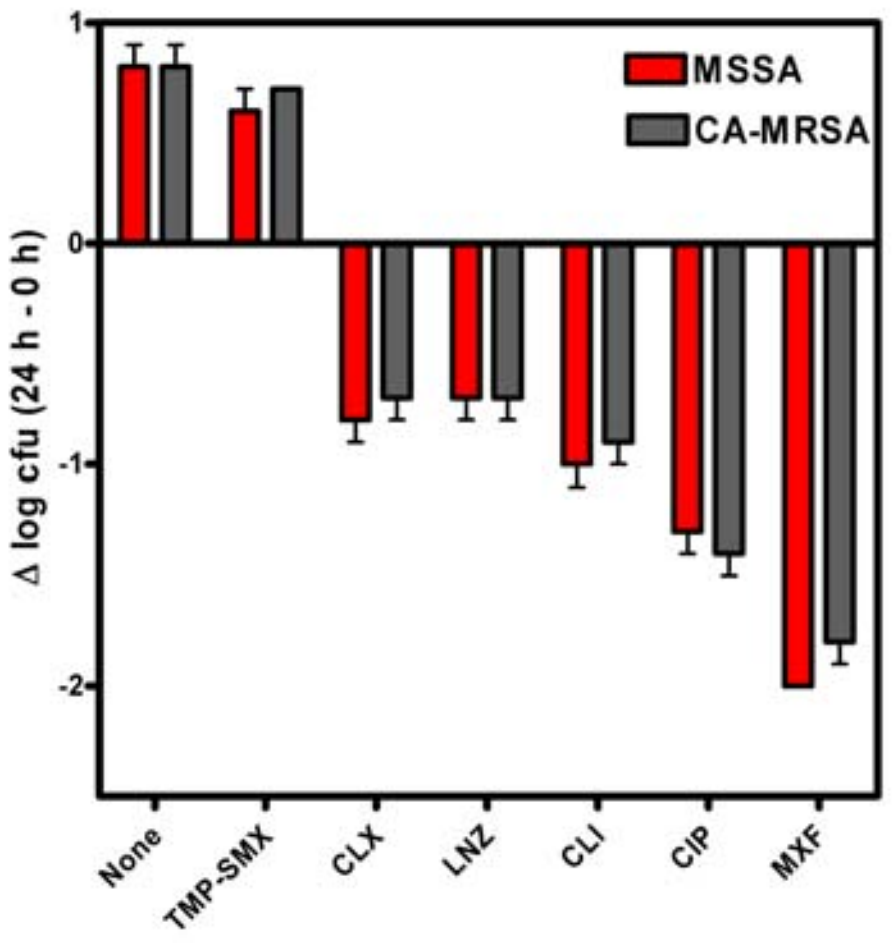

MSSA
CA-MRSA
Δ log cfu (24 h - 0 h)
1
0
-1
-2
None
TMP-SMX
CLX
LNZ
CLI
CIP
MXF

The ordinate shows the change of cfu ($\log_{10}$) per mg of cell protein observed after 24 h of incubation, in comparison with the original inocula (mean ± SD [n=3]).
Cells were incubated with a drug concentration equivalent to the corresponding human Cmax (in mg/L: TMP-SMX [80:16], 25; CLX, 8; LNZ, 21; CLI, 12; CIP, 4; MXF, 4).

Conclusions

- **Among the 6 antibiotics tested, MXF (at clinically achievable concentration) yielded the largest bactericidal effect (~ 2 log cfu decrease) against intracellular *S. aureus*, irrespective of its resistance phenotype towards betalactams;**
- **CLX, LNZ and CLI (commonly recommended for the management of *S. aureus* infections) were less potent (~ 1 log cfu decrease), and TMP-SMX was ineffective in this model.**

Methods

MICs :

Susceptibility testing was performed by micro-dilution method in Mueller-Hinton broth.

Determination of the intracellular antibiotic activity:

THP-1 macrophages were infected with preopsonized bacteria (1 h; 37°C), washed with phosphate-buffered saline, and incubated for 45 minutes with gentamicin (50 mg/Liter) to eliminate non-adherent and non-internalized bacteria.

Infected cells were thereafter exposed for 24 h to antibiotics at a concentration corresponding to their plasma C_{max} as observed in patiens treated with conventional dosages of the corresponding antibiotics (control cells were maintained in the continuous presence of gentamicin [0.5 x MIC] to prevent the extracellular growth of bacteria released from dead cells).

The model and its validation are described in details in refs 2 and 4.

References

1. Lowy FD, Trends Microbiol. 2000;8:341-343
2. Barcia-Macay et al., *Antimicrob. Agents Chemother.*, 2006; 50:841-851.
3. Bronner et al., *Antimicrob. Agents Chemother.*, 2003; 47:1621-1629.
4. Lemaire et al., *J Antimicrob Chemother*, 2005;55:897-904.

Acknowledgments

S. Lemaire is *Boursière* of the Belgian *Fonds pour l'Encouragement de la Recherche dans l'Industrie et l'Agriculture* (FRIA)

This work was supported by the Belgian *Fonds de la Recherche Scientifique Médicale* (FRSM) and with a grant-in-aid from Bayer HealthCare AG.

A copy of this poster will be made available after the meeting at http://www.facm.ucl.ac.be/posters.htm